C1q Inhibition Reduces Neurodegenerative Damage and Improves Survival in the HD R6/2 Mouse Model

Christina Huynh[1], Alessia Tassoni, Vidhu Mathur, Joseph Vereen, Logan Kuhn, Sethu Sankaranarayanan, Ellen Cahir-McFarland, Larry Mattheakis, Ted Yednock, Yaisa Andrews-Zwilling

1. Annexon Biosciences, 1400 Sierra Point Pkwy, 2nd floor, Brisbane, CA 94005

ANNEXON biosciences

BACKGROUND

Huntington's disease (HD) is an autosomal dominant neurodegenerative disease characterized by progressive movement disorder, dementia, and psychosis. It is caused by the increased CAG repeats in the Huntingtin (HTT) gene that results in toxic protein aggregates in neurons and subsequent neuronal death.

Increased expression of early classical complement components, including C1q, C4 and C3, have been observed in striatal tissue from HD patients (Singhrao SK, et al. *Exp Neurol.* 1999). C1q has been implicated in synapse elimination and neuronal damage in HD mouse models with increased neurofilament light-chain (NfL) levels, however, efficacy of complement-based therapy has not been examined. To test the role of the classical complement pathway in neuronal damage, we used the R6/2 transgenic mouse model of HD and pharmacologically blocked C1q activity with an inhibitory antibody (ANX-M1), administered via intraperitoneal injection, and assessed NfL, synaptic and neuronal changes, microglia reactivity and animal survival.

OBJECTIVE

To examine complement expression, neuronal damage, and the potential therapeutic benefit of classical complement inhibition with anti-C1q antibody (Annexon Biosciences) in the transgenic R6/2 mouse model of HD.

METHOD

R6/2 mice (hemizygous, 120 CAG repeats, Jackson Laboratory) and their littermate controls were dosed twice per week intraperitoneally (IP) with either anti-C1q antibody (ANX-M1), isotype control, or received no treatment (Table 1). We assessed longitudinal changes in NfL levels in cerebral spinal fluid (CSF) and plasma. Complement components were measured in plasma and brain lysates from animals at 6, 10, and 15 weeks of age.

Levels of NfL were measured in CSF and plasma at Quanterix using Single Molecule Array (SIMOA) Technology. Complement signature in plasma and brain protein lysate was measured using standard sandwich ELISA based assays. Levels of Iba1, CD68, NeuN and FluoroJade C labelling were assessed by immunofluorescence. Western Blot was used to measure levels of the pre-synaptic marker VGLUT1.

Table 1. Study Design

Group	Number	Genotype	Treatment	Route	Frequency of Administration	Duration of Treatment
1	18	Hemizygous (R6/2)	mIgG1 Isotype Control at 100 mg/mL	IP (16 mL/kg)	Twice per week	~4.5W - 13W
2	18	Hemizygous (R6/2)	ANX-M1 at 100 mg/mL	IP (16 mL/kg)	Twice per week	~4.5W - 13W
3	10	WT	NT			

RESULTS

Figure 1. Increased Level of NfL, a Biomarker of Neuronal Damage, in CSF and Plasma

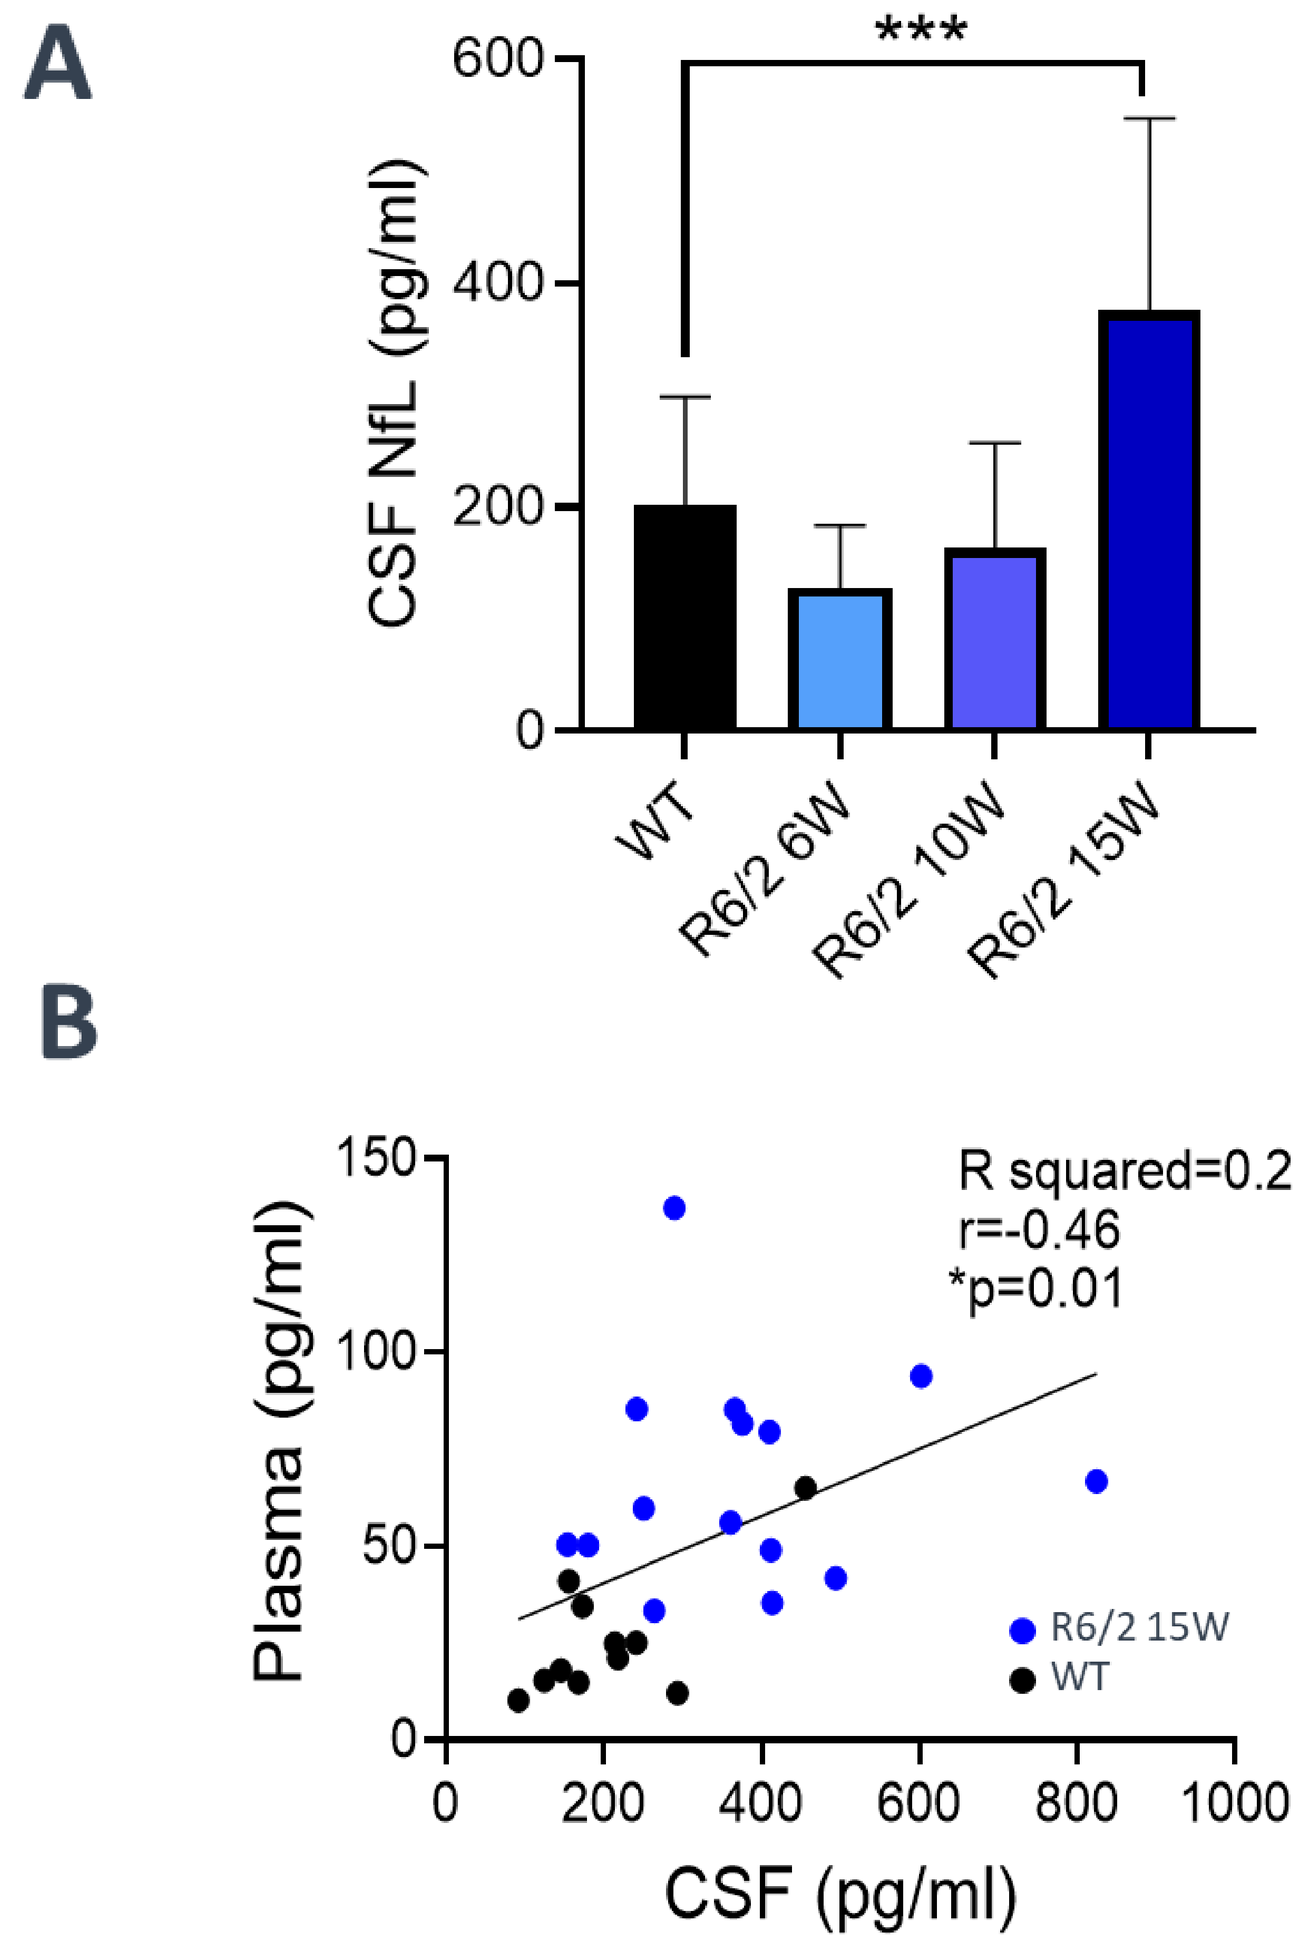
A
CSF NfL (pg/ml)
WT
R6/2 6W
R6/2 10W
R6/2 15W
B
R squared=0.2
r=-0.46
*p=0.01
Plasma (pg/ml)
CSF (pg/ml)
R6/2 15W
WT

Longitudinal levels of NfL measured in CSF of R6/2 mice and WT littermates. (A) NfL significantly increased as disease progressed in CSF compared to WT at 14 weeks of age (***p≤0.001). (B) Correlation analysis between CSF and plasma NfL levels at 15 weeks of age (Pearson r=0.46; *p=0.01).

Figure 2. Changes in Complement Expression and Activation in Brain and Plasma with Disease Progression

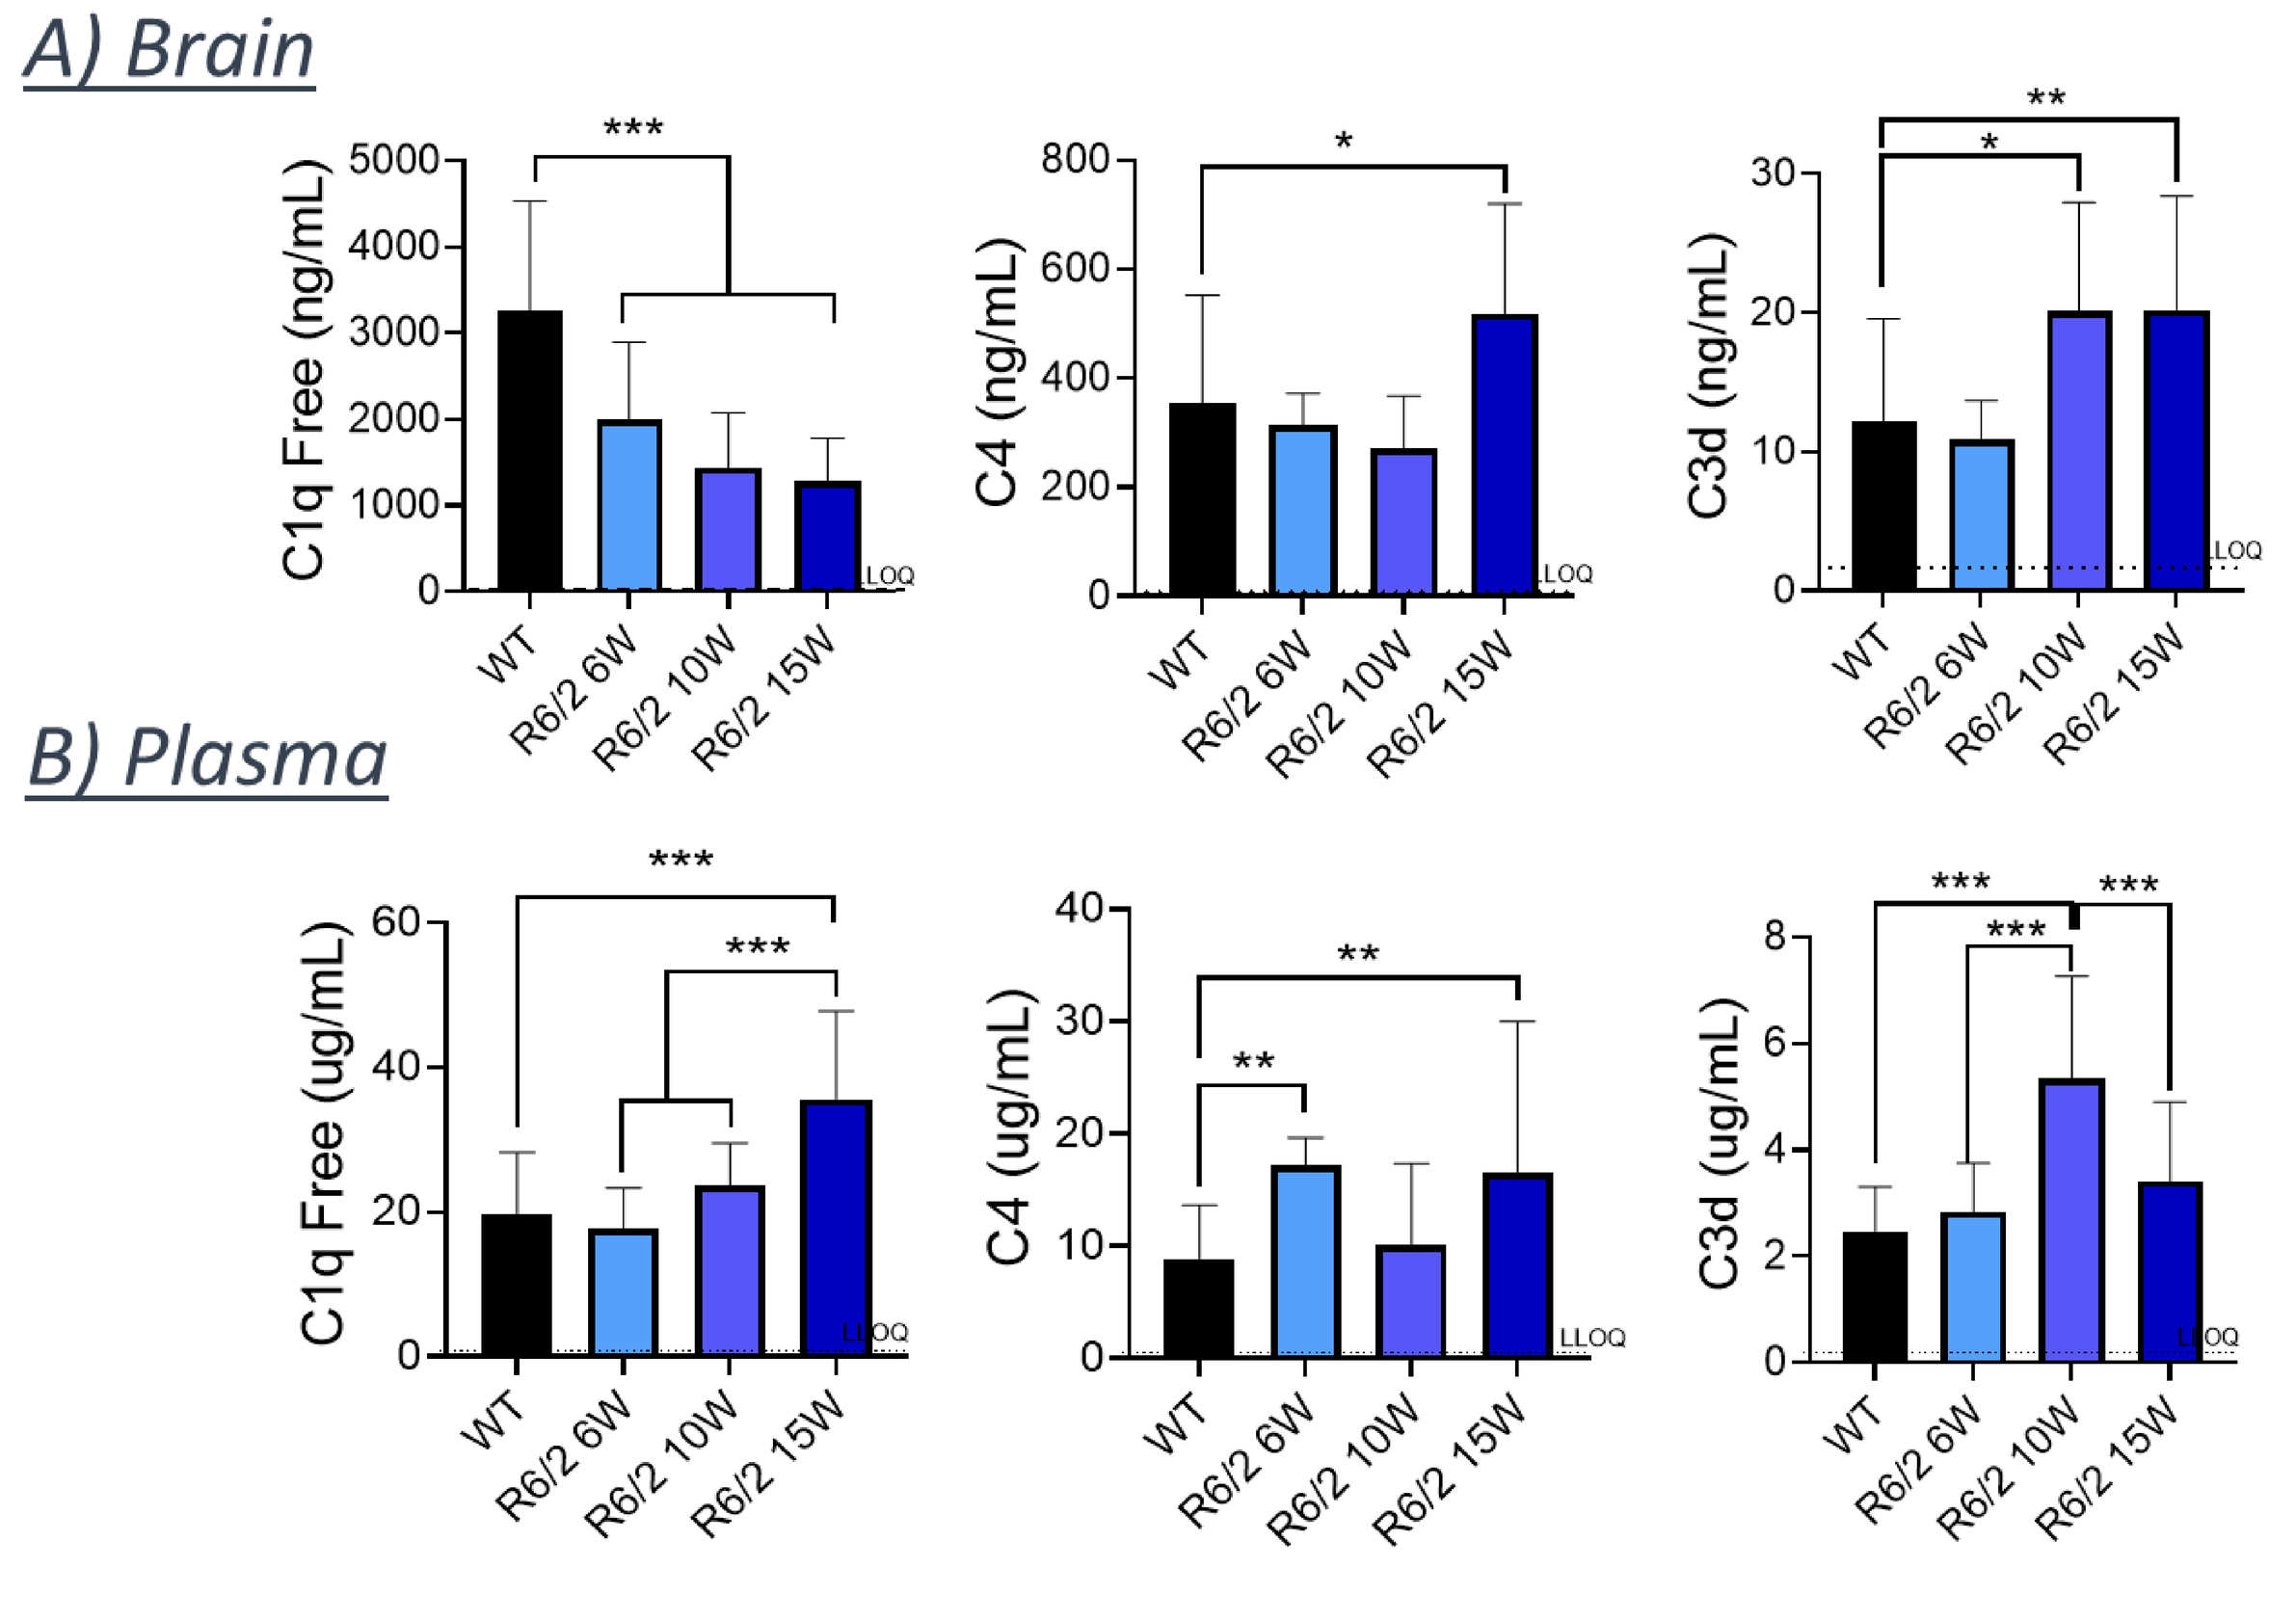
A) Brain
C1q Free (ng/mL)
C4 (ng/mL)
C3d (ng/mL)
B) Plasma
C1q Free (ug/mL)
C4 (ug/mL)
C3d (ug/mL)
WT
R6/2 6W
R6/2 10W
R6/2 15W
LLOQ

Longitudinal measures of complement proteins in (A) brain lysates and (B) plasma. Significant increase of C1q, C4, and C3d levels observed with disease progression. (*p≤0.05; **p≤0.01; ***p≤0.001)

Figure 3. Anti-C1q Treatment Significantly Inhibited the Classical Complement Pathway in Brain and Plasma

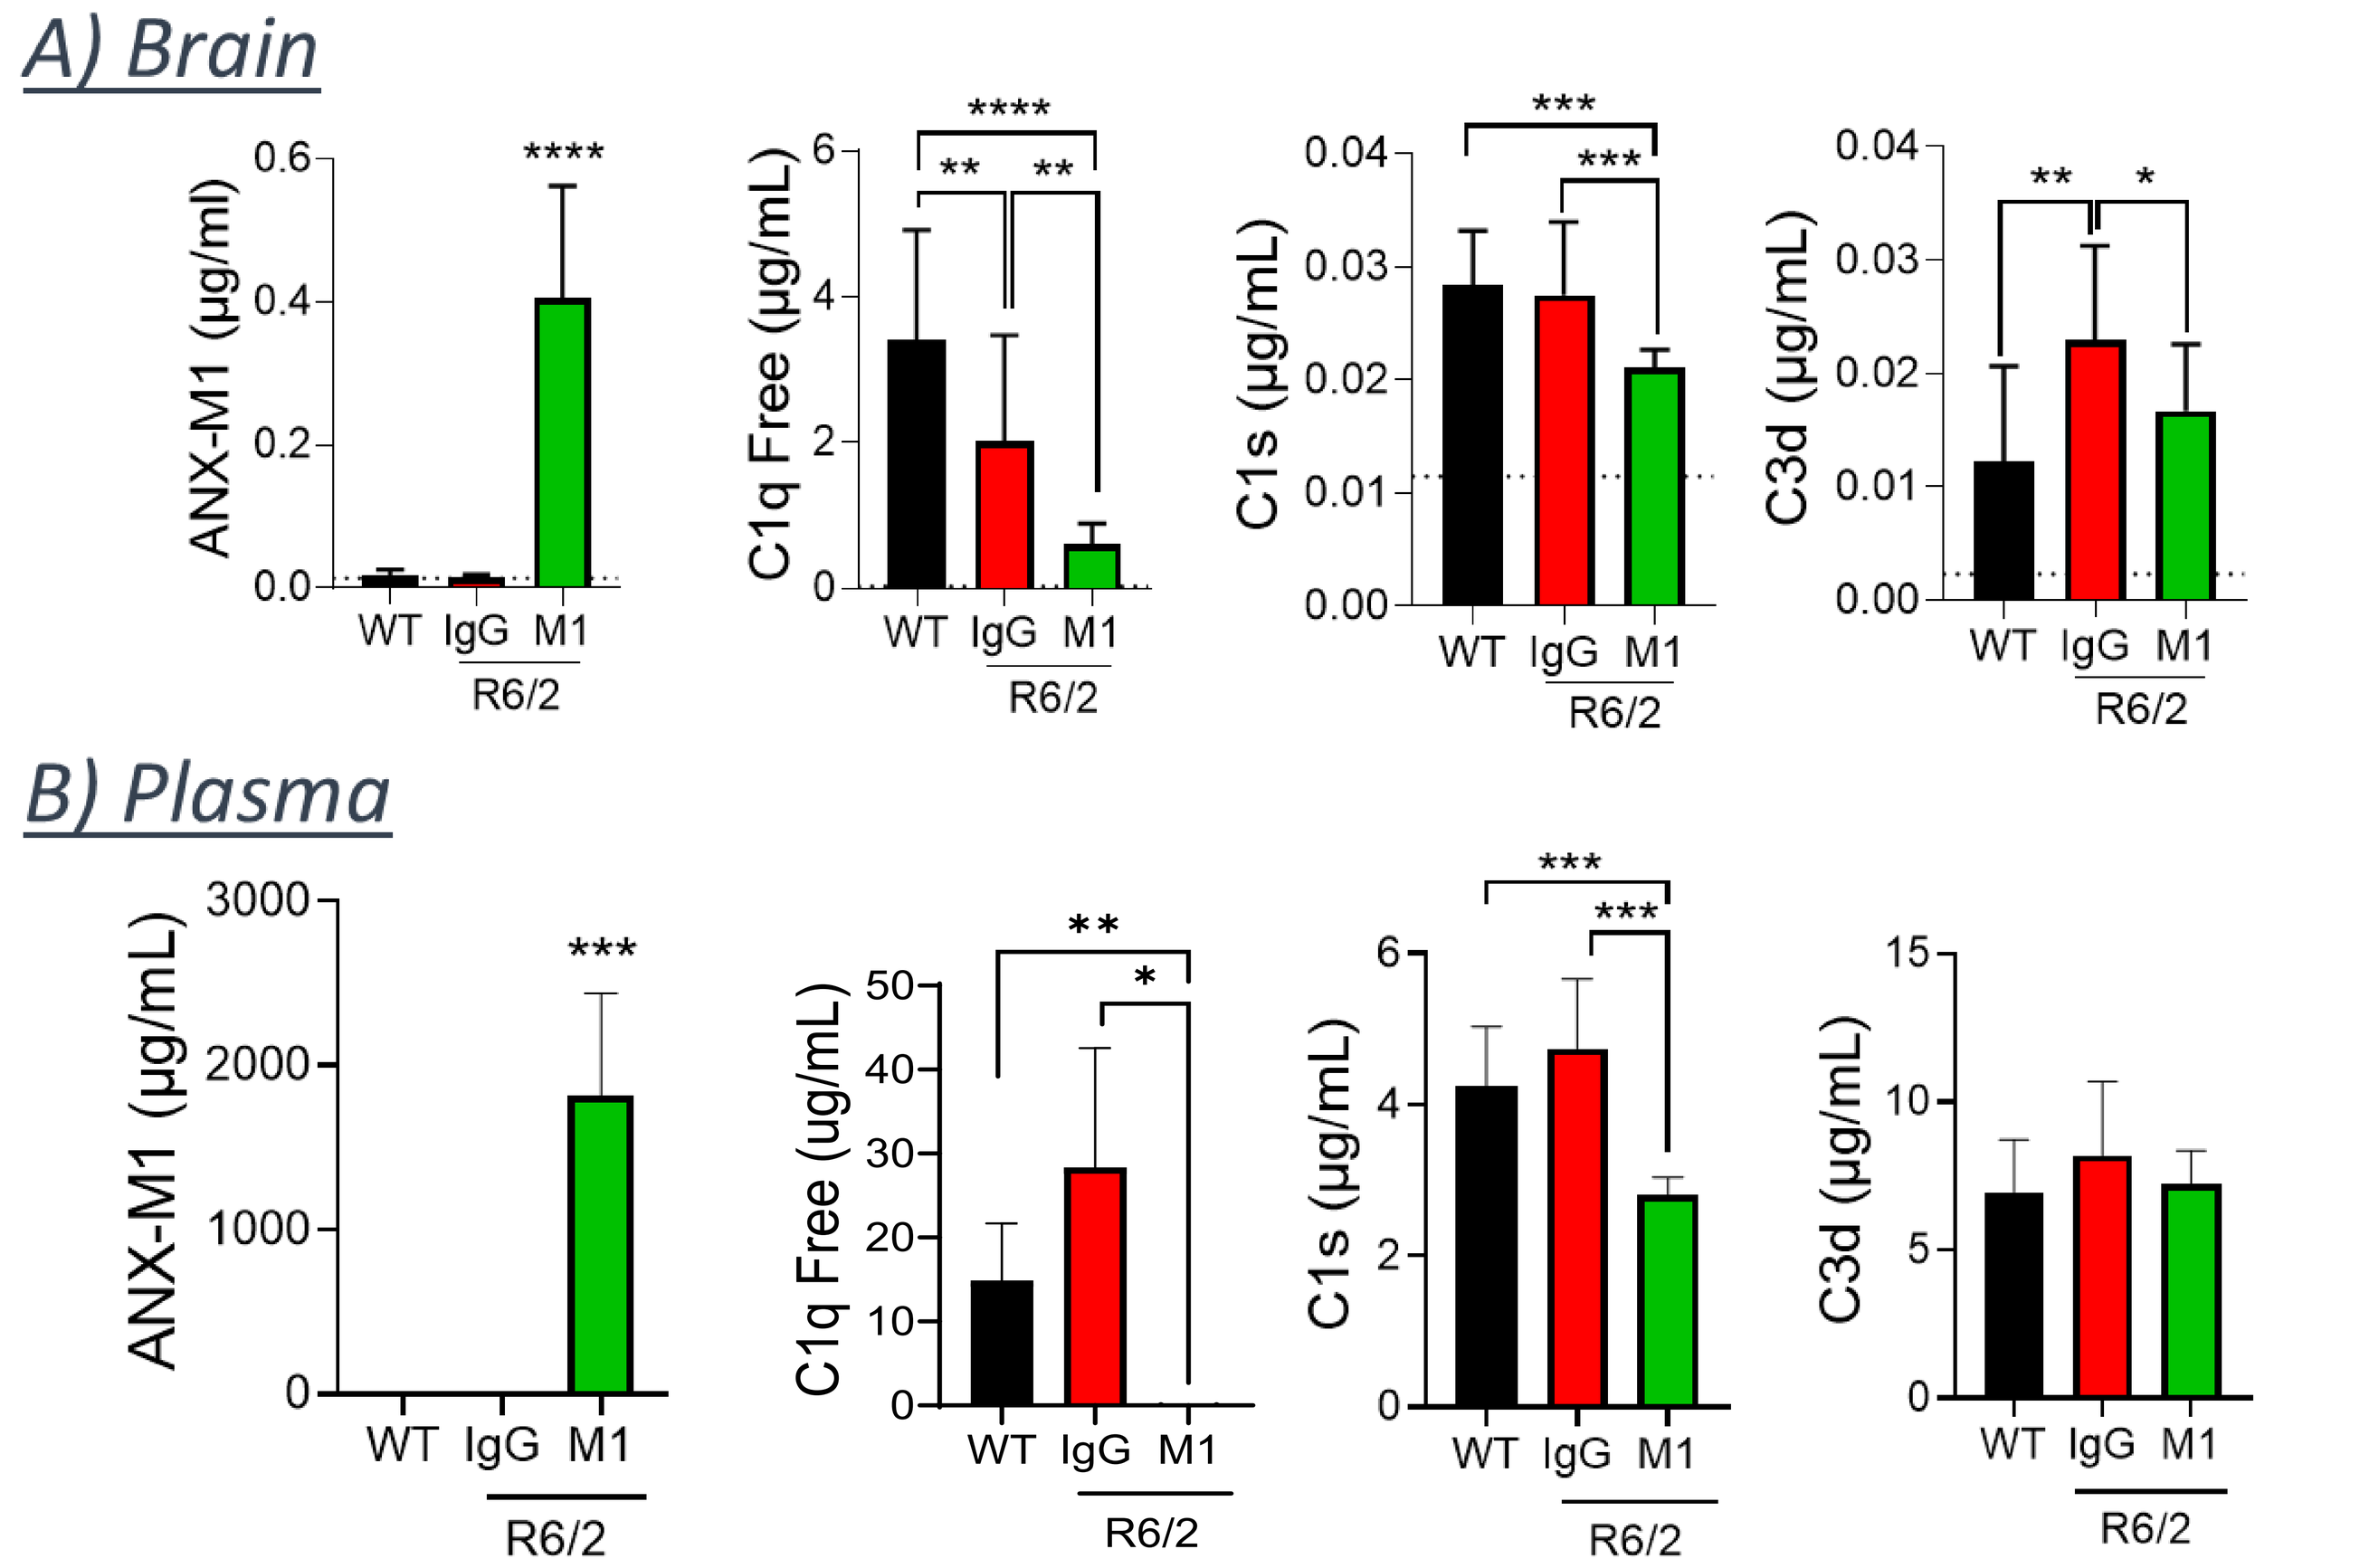
A) Brain
ANX-M1 (µg/ml)
C1q Free (µg/mL)
C1s (µg/mL)
C3d (µg/mL)
B) Plasma
ANX-M1 (µg/mL)
C1q Free (ug/mL)
C1s (µg/mL)
C3d (µg/mL)
WT
IgG
M1
R6/2

PK/PD analysis shows detectable drug levels in R6/2 mouse (A) brains (*p and (B) plasma. Significant decrease in complement levels of C1q, C1s, and C3d observed in both brain and plasma upon anti-C1q treatment, confirming target engagement. (*p≤0.05; **p≤0.01; ***p≤0.001)

Figure 4. Systemic Anti-C1q Treatment Reduces Microglia Reactivity in the Dorsal Striatum

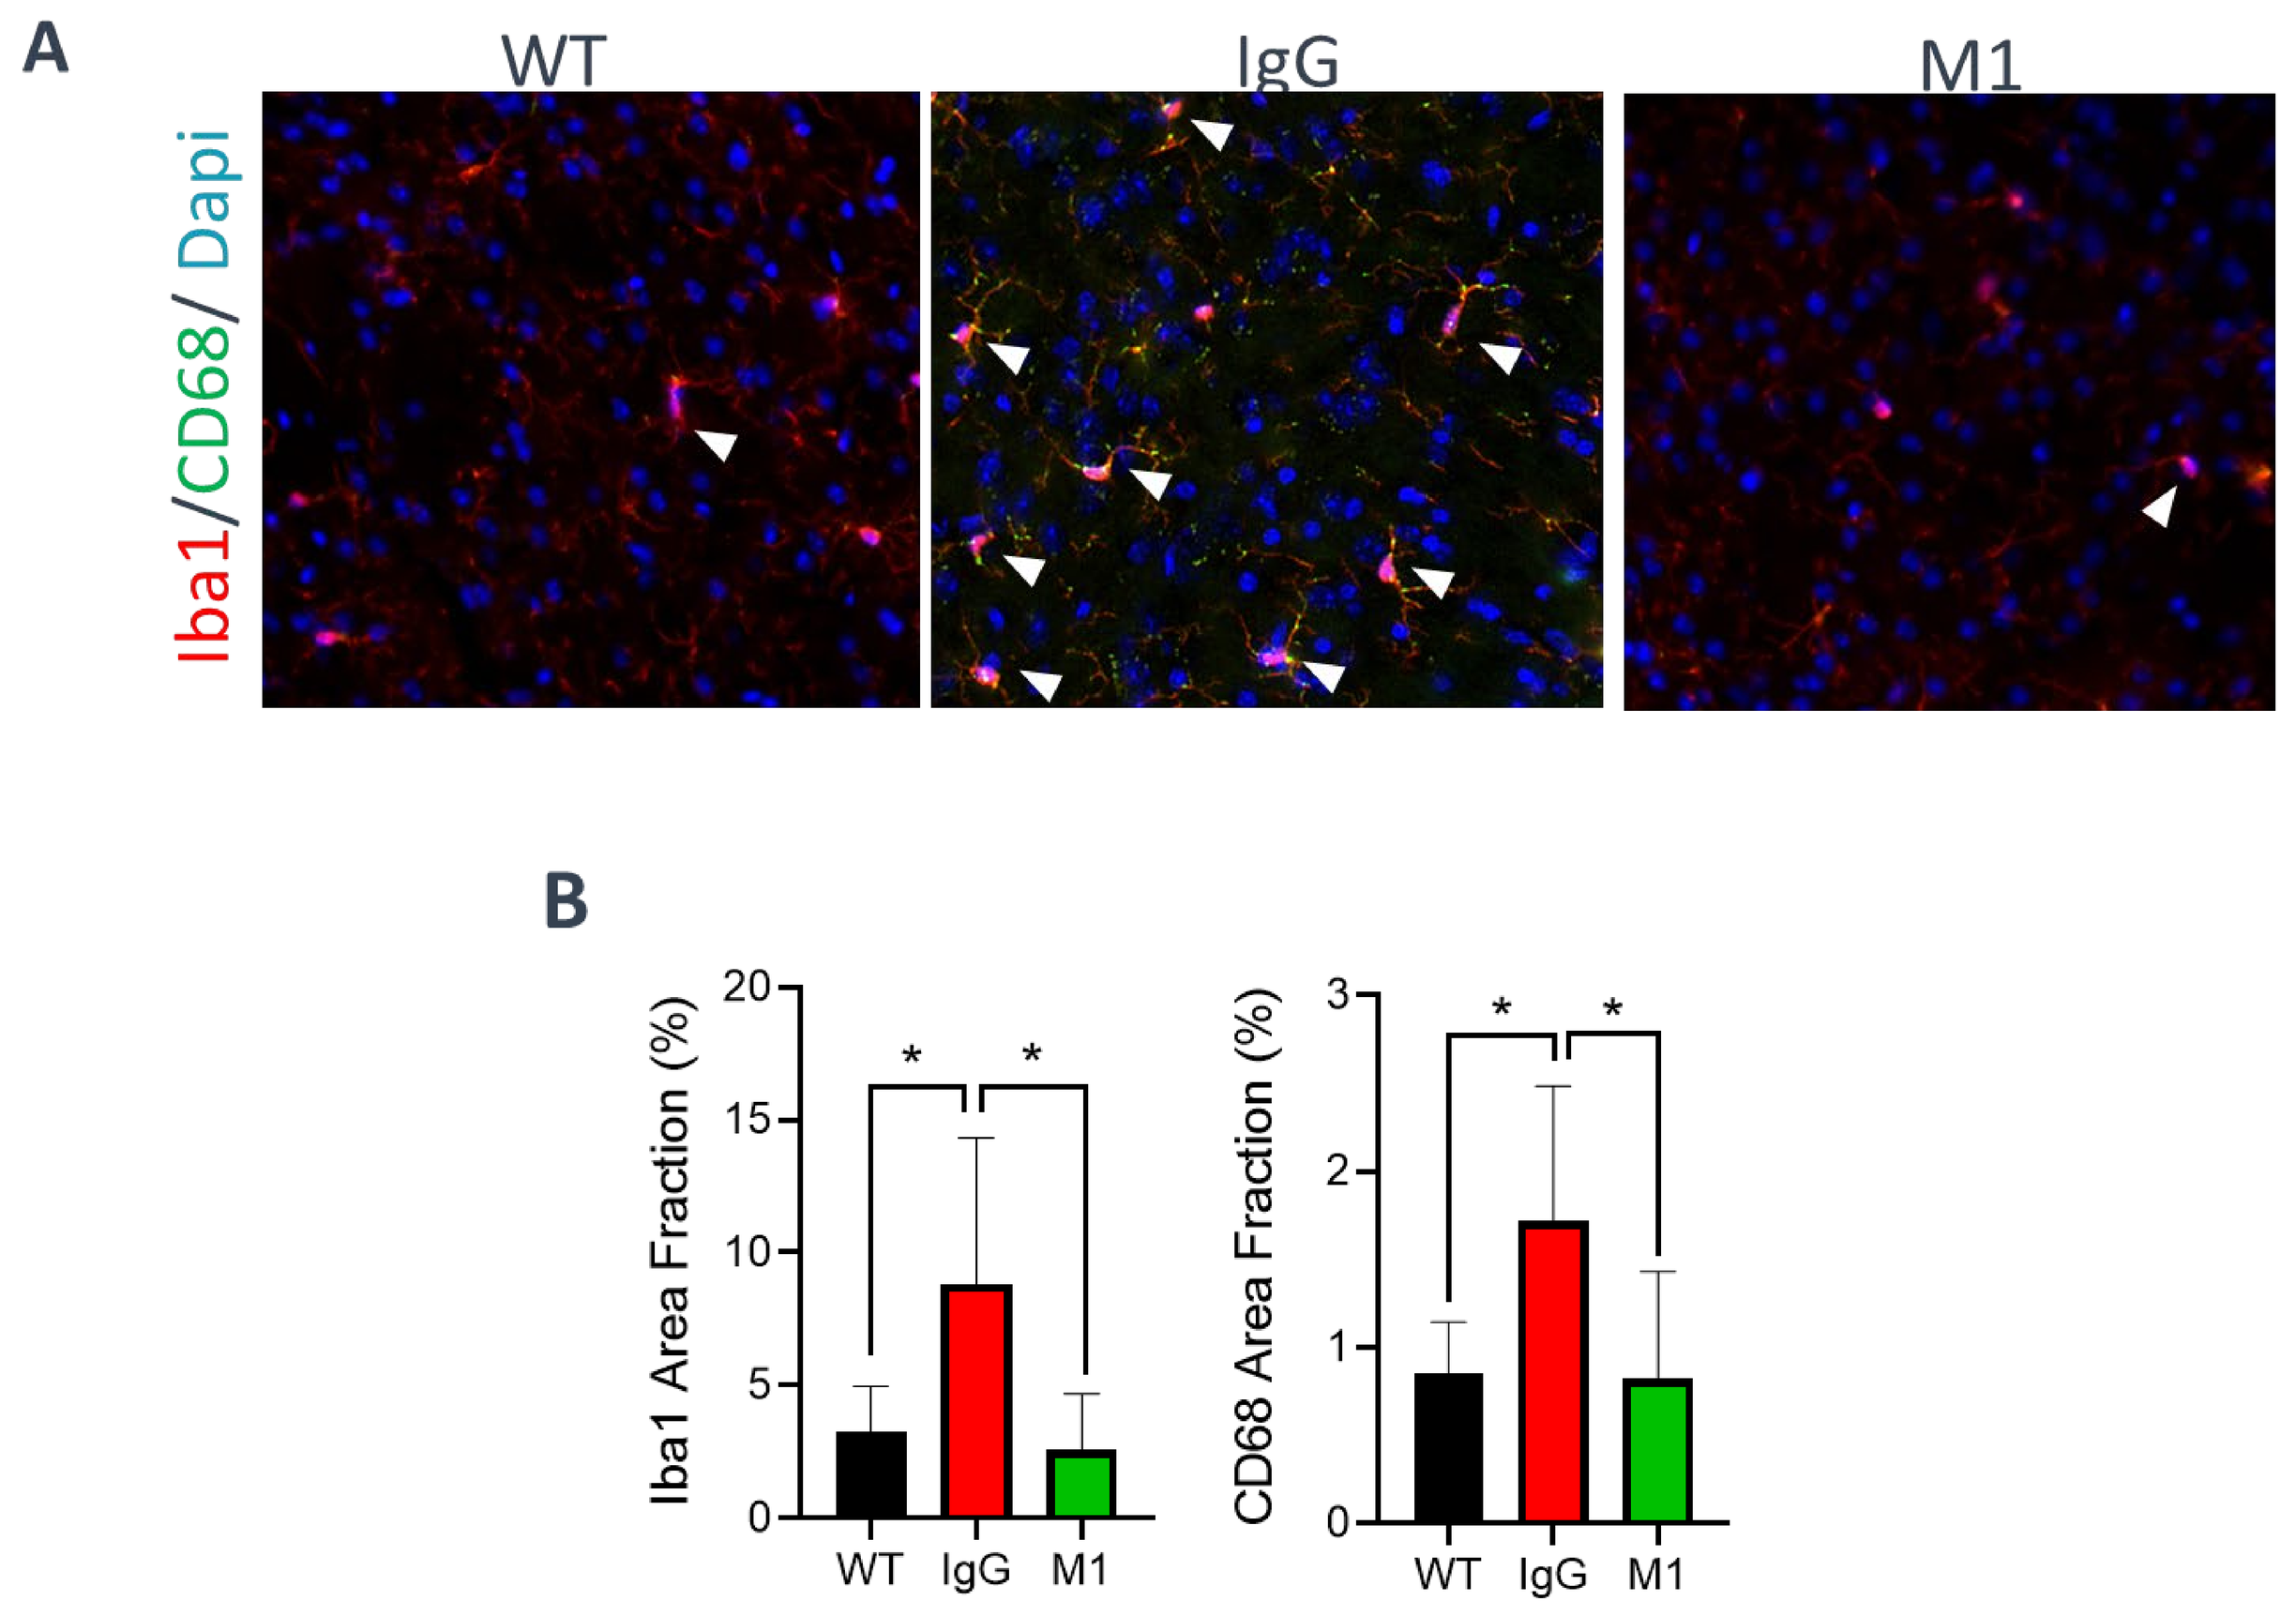
A
WT
IgG
M1
Iba1/CD68/ Dapi
B
Iba1 Area Fraction (%)
CD68 Area Fraction (%)
WT
IgG
M1

(A) Immunofluorescence (IF) labeling of dorsal lateral striatal brain sections for pan-microglia marker (Iba1, red) and phagocytic marker (CD68, green). Arrowheads point to colocalizations of the markers. (B) Quantification analysis shows reduction in microglia reactivity upon anti-C1q treatment. (*p≤0.05)

Figure 5. Anti-C1q Treatment Reduces Neurodegeneration

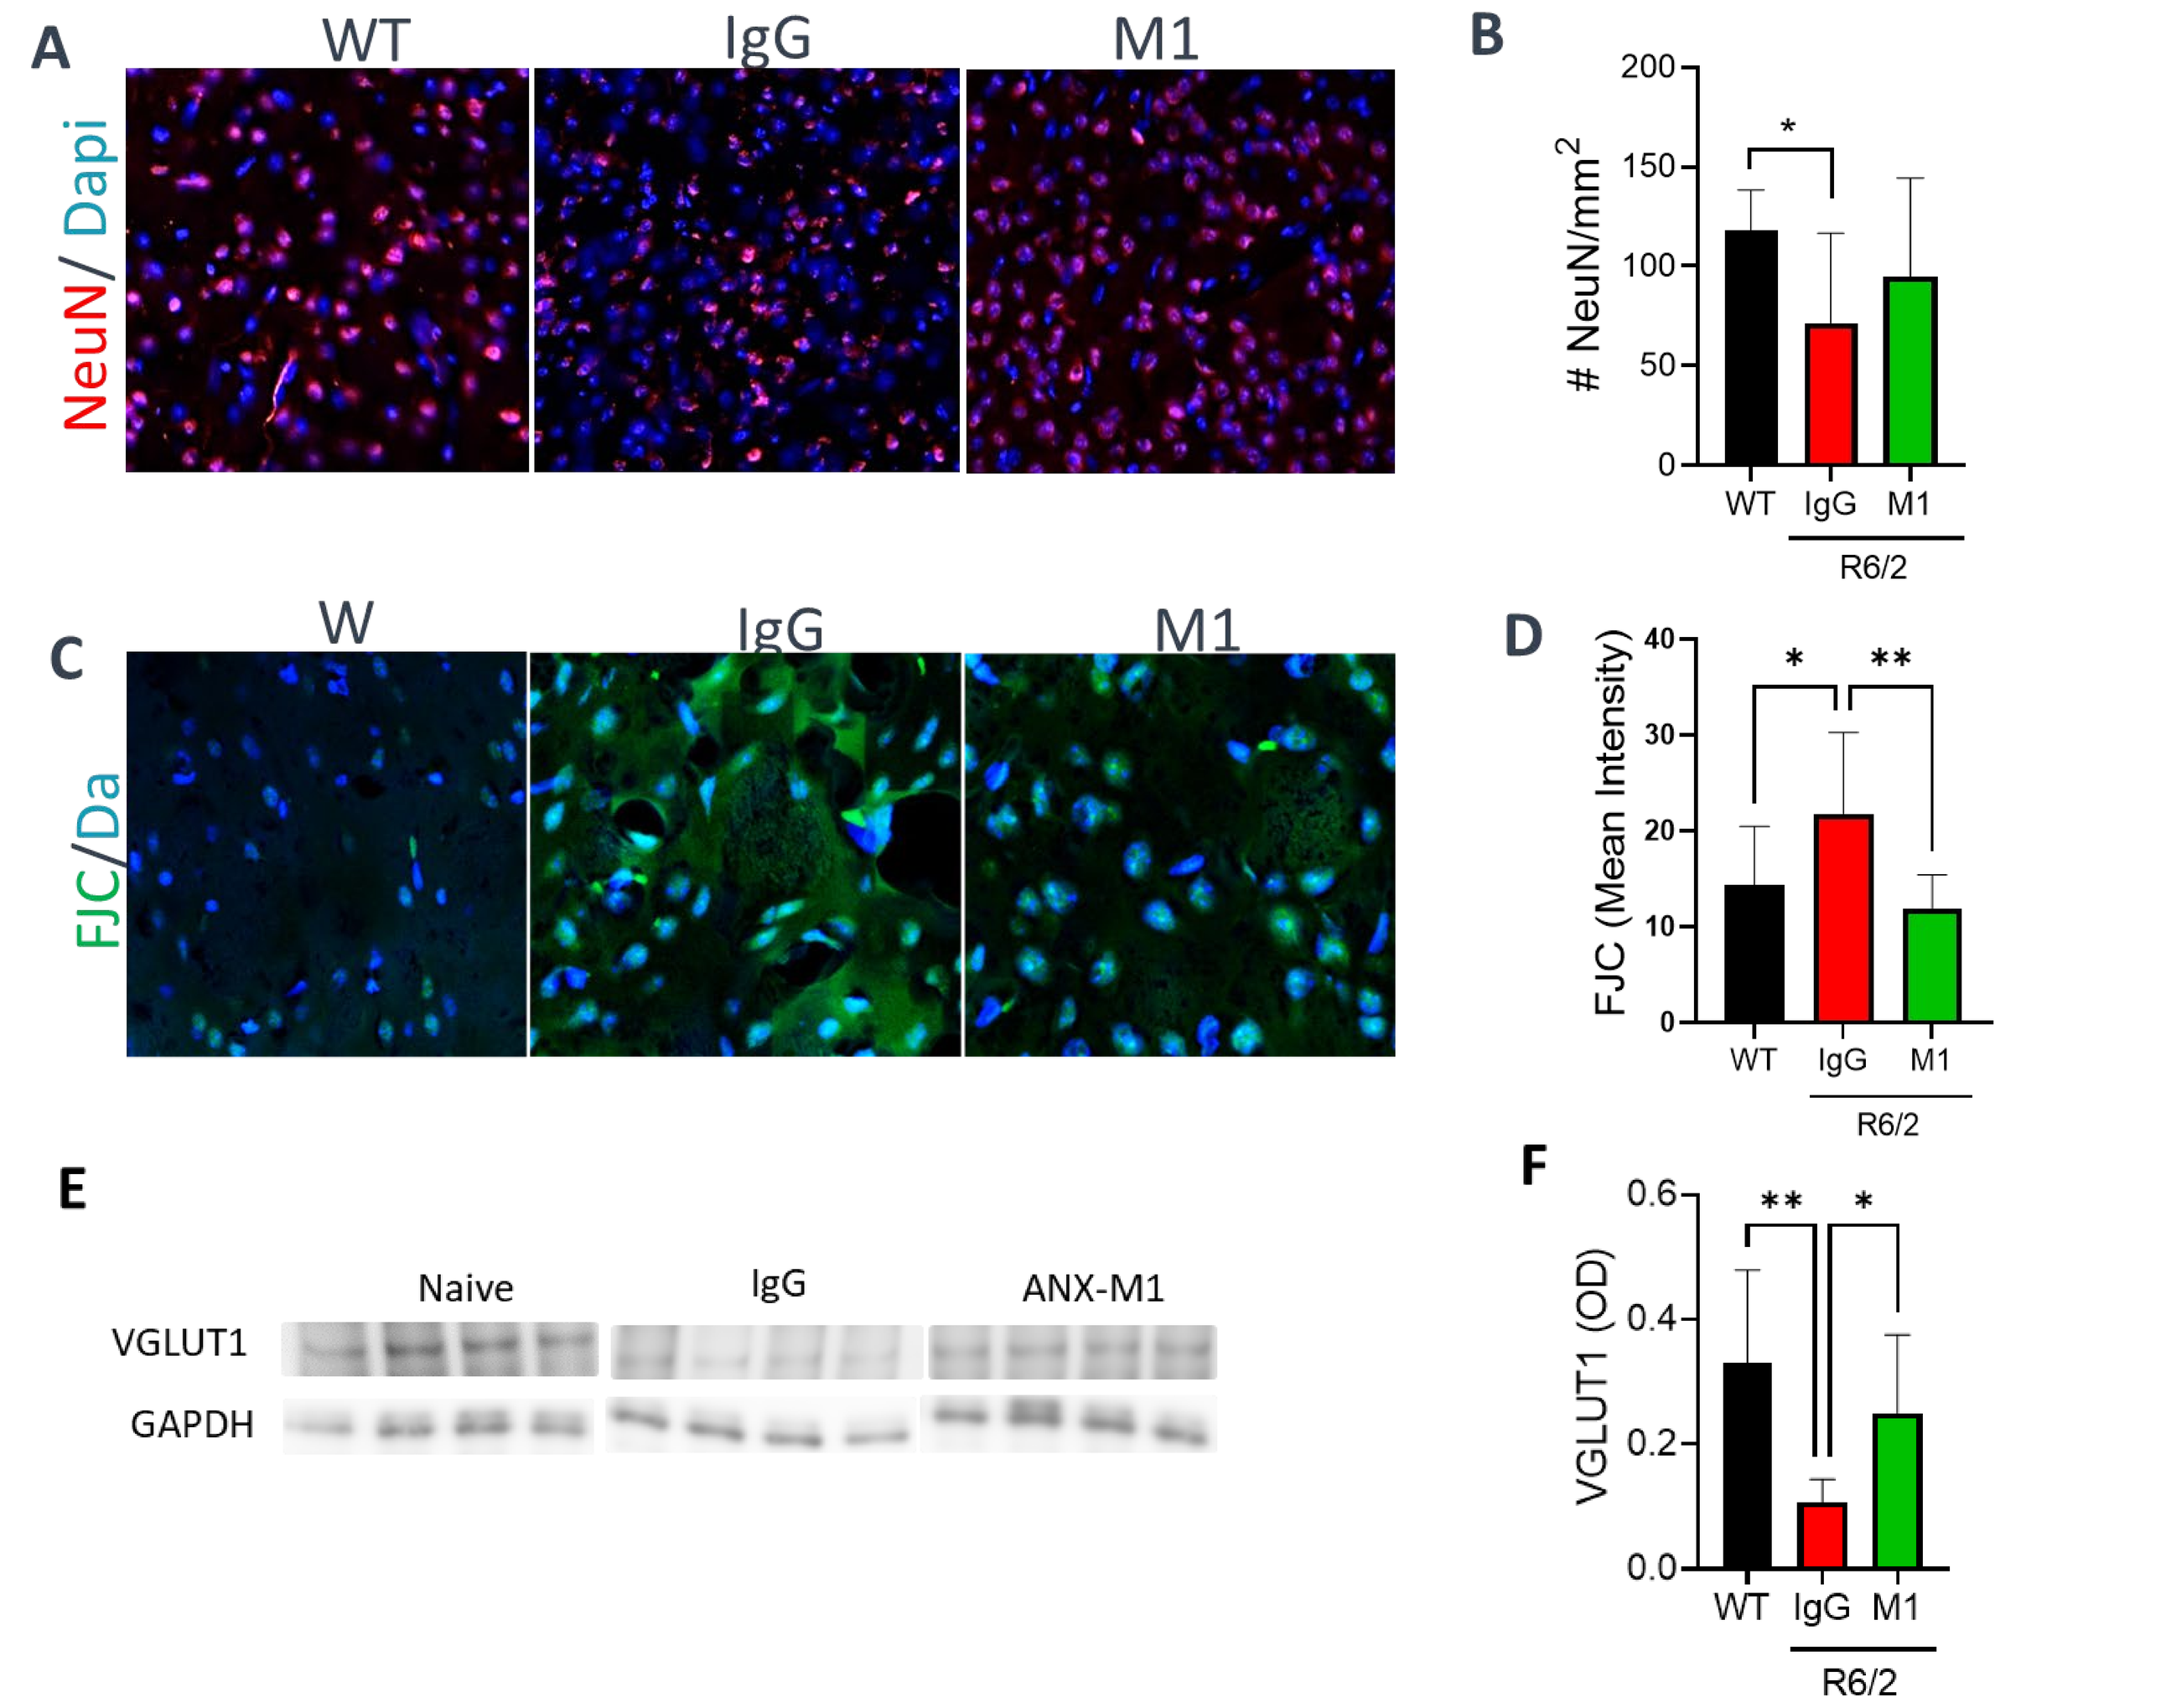
A
WT
IgG
M1
NeuN/ Dapi
B
NeuN/mm2
C
W
IgG
M1
FJC/Da
D
FJC (Mean Intensity)
E
Naive
IgG
ANX-M1
VGLUT1
GAPDH
F
VGLUT1 (OD)
R6/2

Although no significant changes were observed in total neuron count (NeuN, red; A and B) there was significant reduction in the number of degenerating neurons as measured with Fluoro-Jade-C (green; C and D), suggesting a neuroprotective effect with anti-C1q treatment. Consistent with this result, western blot analysis of the pre-synaptic marker (VGLUT1) in brain lysates indicates significant synapse preservation with anti-C1q treatment. (E and F; *p≤0.05; **p≤0.01)

Figure 6. Systemic Anti-C1q Treatment Reduces CSF-NfL Levels and Improves Survival

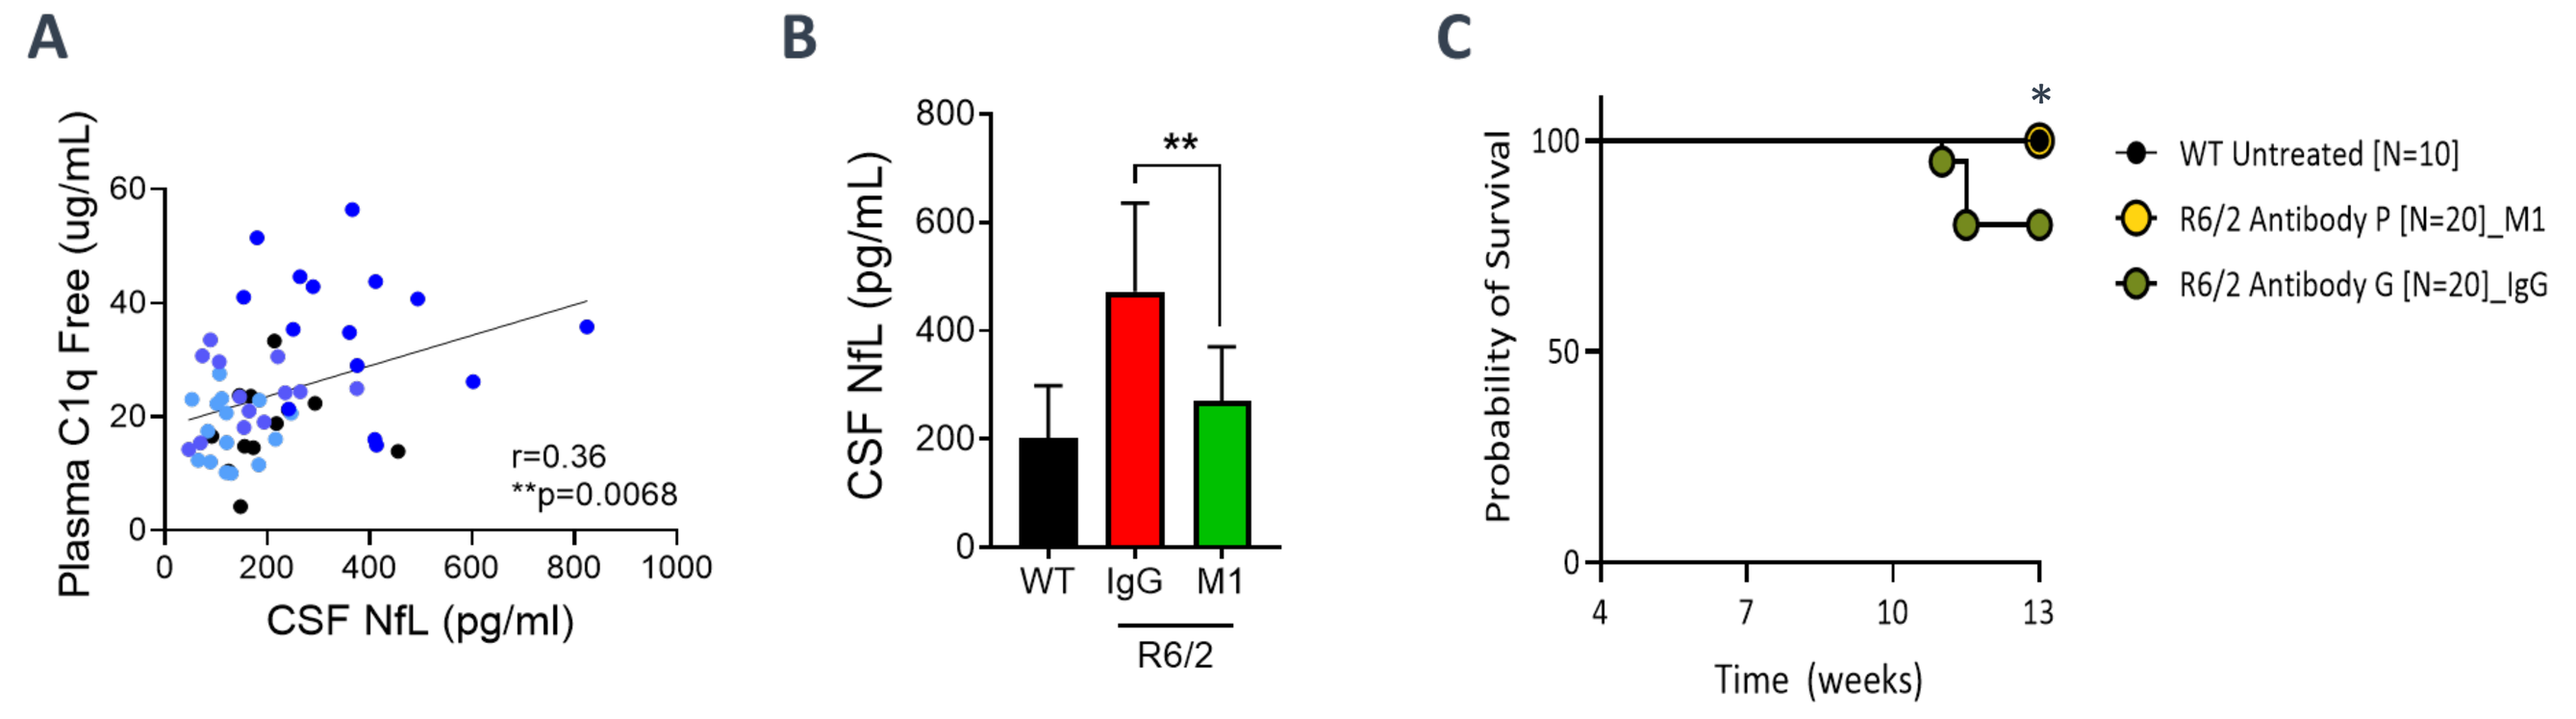
A
Plasma C1q Free (ug/mL)
CSF NfL (pg/ml)
r=0.36
**p=0.0068
B
CSF NfL (pg/mL)
WT
IgG
M1
R6/2
C
Probability of Survival
Time (weeks)
WT Untreated [N=10]
R6/2 Antibody P [N=20]_M1
R6/2 Antibody G [N=20]_IgG

(A) Significant positive correlation between plasma C1q and CSF-NfL levels with disease progression (Pearson r=-0.36, **p=0.006). (B) Reduced level of NfL in R6/2 mice upon anti-C1q treatment consistent with neuroprotection (**p≤0.01). (C) Kaplan Meier survival curve shows signal of increased survival observed in R6/2 mice following treatment (*p=0.037).

CONCLUSIONS

- Inhibiting C1q protects against neuronal damage in R6/2 Huntington's mice
- Classical complement component C1q represents a potential pharmacological target in HD
- A Phase 2 study of ANX005, an anti-C1q therapy, in patients with HD was completed this year (clinicaltrials.gov NCT04514367)

Society for Neuroscience 2022

#534.18