C1q Inhibition Reduces Neurodegenerative Damage, Preserves Neuromuscular Junctions and Improves Compound Muscle Action Potential in the SOD1^{G93A} Model

Joseph Vereen, Alessia Tassoni, Vidhu Mathur, Christina Huynh, Ellen Cahir-McFarland, Larry Mattheakis, Ted Yednock, Yaisa Andrews-Zwilling

Annexon Biosciences, 1400 Sierra Point Pkwy, 2nd floor, Brisbane, CA 94080

ANNEXON biosciences

Abstract

Amyotrophic lateral sclerosis (ALS) is a sporadic or genetic disease associated with peripheral loss of synaptic connectivity at the neuromuscular junction (NMJ) and central loss of motor neurons. The SOD1^{G93A} mouse model has been widely used to study a familial form of ALS. C1q, the initiating molecule of the classical complement cascade, marks synapses in the central nervous system for glial elimination during normal development, but it also triggers aberrant synapse loss in neurodegenerative disorders. In ALS, C1q also tags the NMJ within the peripheral nervous system prior to its removal by macrophages. We hypothesized that excessive synaptic pruning initiated by C1q contributes to motor deficits in ALS and that pharmacologically inhibiting this process would be beneficial. We treated adult SOD1^{G93A} mice with a C1q-blocking antibody (anti-C1q) from 7 to 16 weeks of age. Treatment for 9 weeks resulted in reduction in C1q level in plasma and spinal cord and muscle tissue, along with inhibition of downstream classical complement activation. Treated mice showed significant preservation of NMJ density, as measured by bungarotoxin labelling of the gastrocnemius muscle, and improvement in the amplitude of compound muscle action potential, demonstrating that C1q inhibition leads to increased synaptic connectivity at the NMJ. Furthermore, anti-C1q treatment reduced Nf-L levels in the cerebrospinal fluid and plasma of SOD1^{G93A} mice compared to those observed in untreated mice, indicating reduced neuronal damage in this model. These findings suggest that inhibition of the classical complement pathway results in reduced Nf-L levels, NMJ preservation, and improved muscle nerve conduction following anti-C1q therapy treatment in the SOD1^{G93A} mouse model of ALS. A Phase 2 study of ANX005, an anti-C1q therapy, in ALS patients is ongoing.

Hypothesis

C1q contributes to neuronal damage and loss of motor function in the SOD1^{G93A} mouse model of ALS by tagging neuromuscular junctions (NMJ) and driving complement dependent synaptic pruning by macrophages.
We evaluated the hypothesis that systemic blockade of C1q using an anti-C1q antibody preserves NMJ and slow down disease progression.

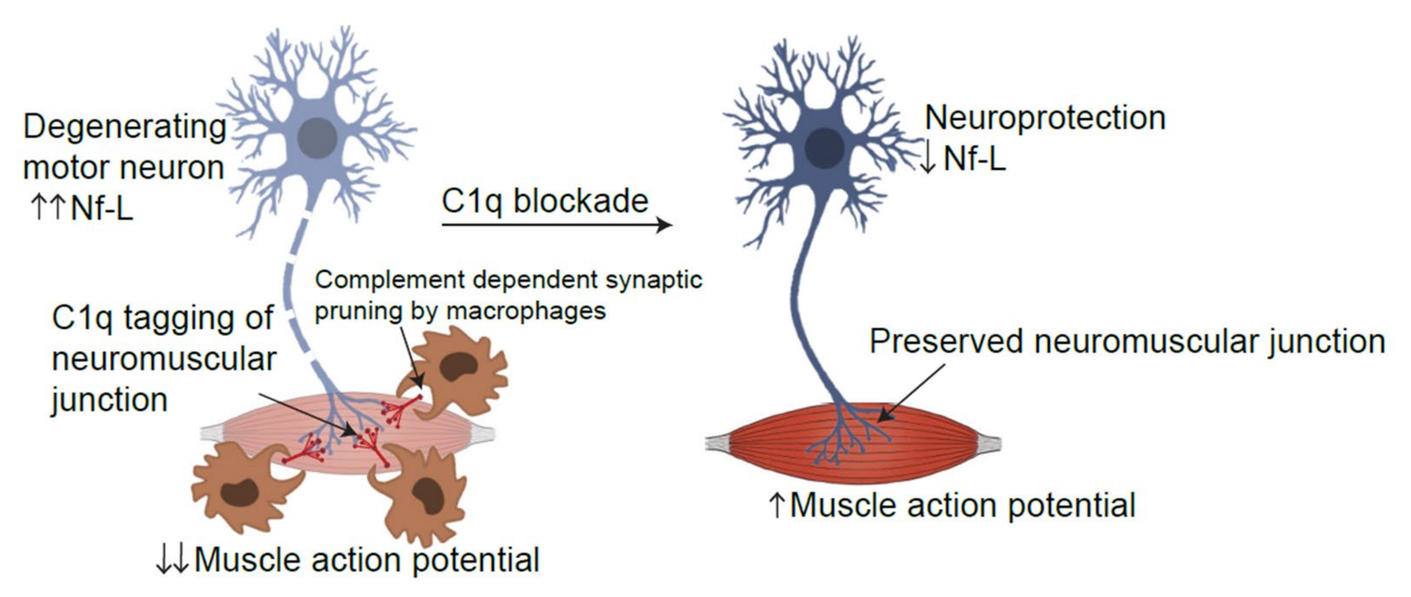

Degenerating motor neuron
↑↑Nf-L
C1q blockade
Neuroprotection
↓Nf-L
Complement dependent synaptic pruning by macrophages
C1q tagging of neuromuscular junction
Preserved neuromuscular junction
↓↓Muscle action potential
↑Muscle action potential

Study Design

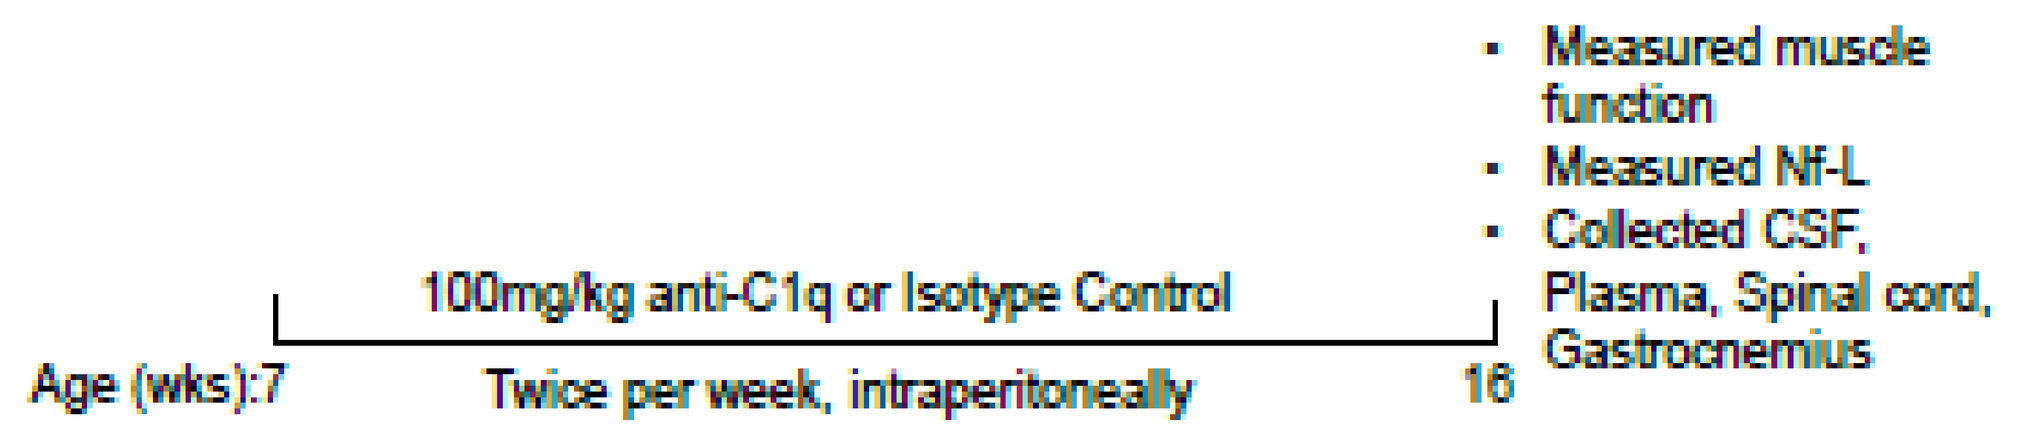

Measured muscle function
Measured Nf-L
Collected CSF, Plasma, Spinal cord, Gastrocnemius
100mg/kg anti-C1q or Isotype Control
Age (wks):7
Twice per week, intraperitoneally
16

SOD1^{G93A} mice were dosed twice per week intraperitoneally with 100mg/kg anti-C1q antibody (Annexon Biosciences) or isotype control starting at 7 weeks till 16 weeks of age (n=16-17/group). Wildtype (WT) mice (n=17) were used as baseline controls. Compound action muscle potential was measured at age 16 weeks. Mice were perfused and CSF, plasma, spinal cord and gastrocnemius muscles were collected 3 days after last dose. In vivo animal work was done at Psychogenics Inc. CSF and plasma Nf-L were measured at using Single Molecule Array (SIMOA) platform.

Results

Figure 1. NMJ Density Correlates with Muscle Function in SOD1^{G93A} Mouse Model

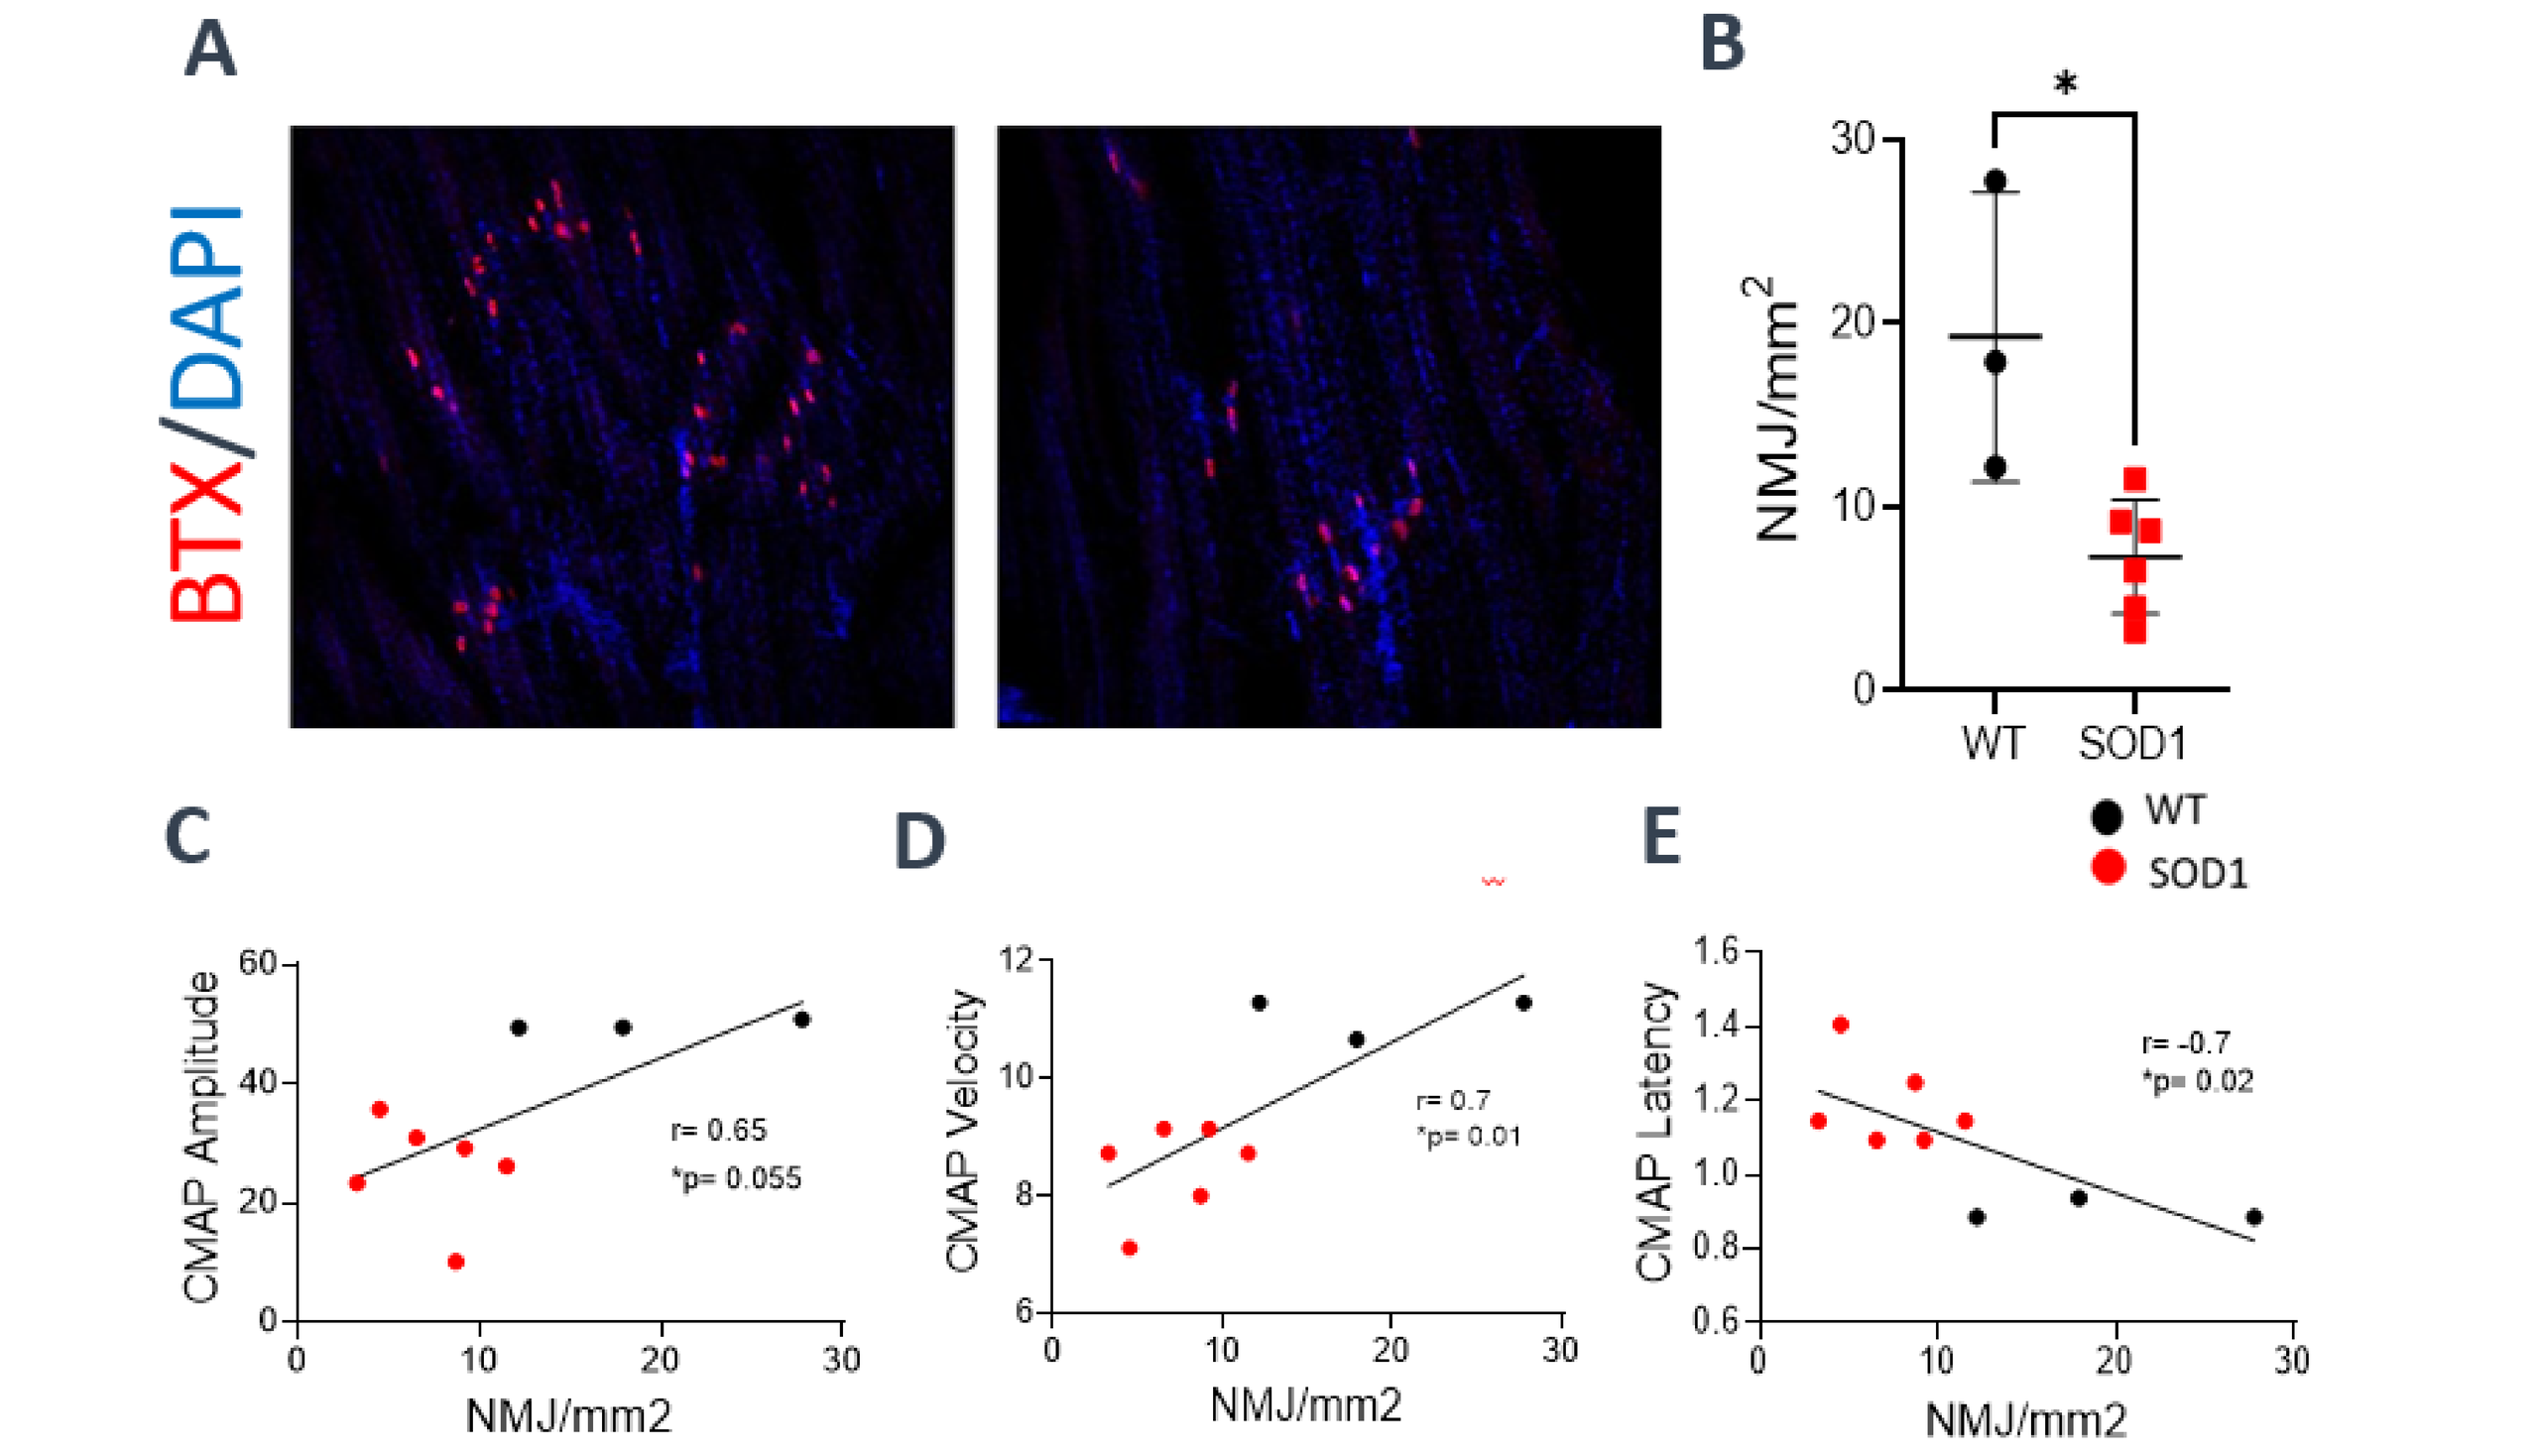

A
BTX/DAPI
B
NMJ/mm2
WT
SOD1
C
CMAP Amplitude
r= 0.65
*p= 0.055
D
CMAP Velocity
r= 0.7
*p= 0.01
E
CMAP Latency
r= -0.7
*p= 0.02
NMJ/mm2

(A-B) Immunofluorescence (IF) images and its quantification showing significant decrease in muscle NMJ density in SOD1^{G93A} mouse model at 15 weeks of age compared to WT littermate control; **C-E**) Significant correlation between NMJ density and muscle function (CMAP Amplitude, Velocity and Latency). : One-way ANOVA with repeated measures *P<0.01, **P<0.001, ***P<0.0001.

Figure 2. C1q Deposition at the NMJ and Expression by Infiltrating Macrophages in the SOD1^{G93A} Mouse Model

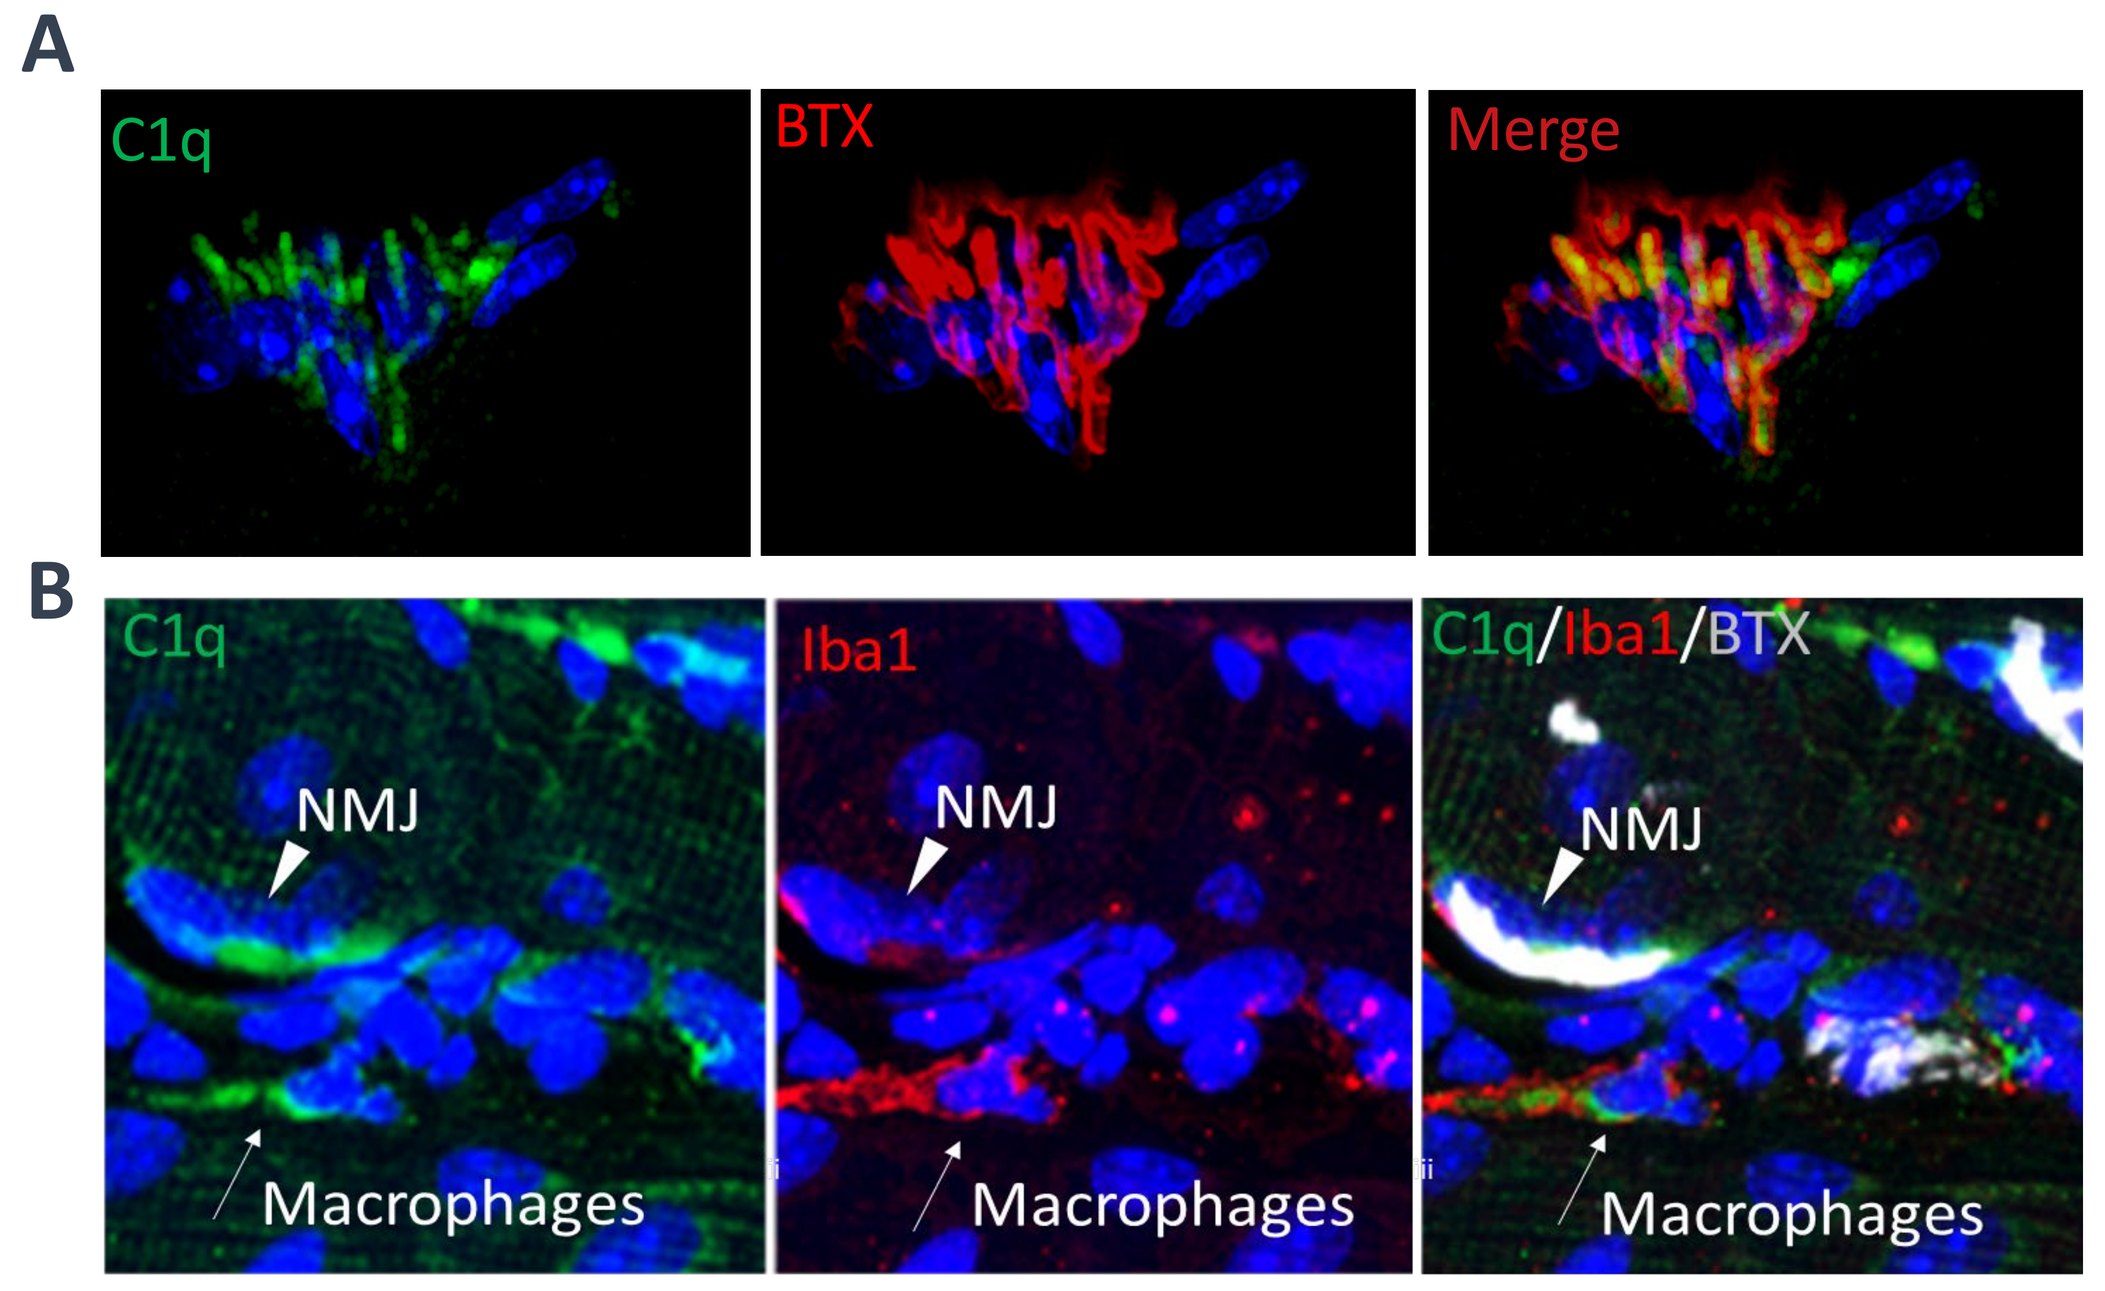

A
C1q
BTX
Merge
B
C1q
Iba1
C1q/Iba1/BTX
NMJ
Macrophages

Immunofluorescent images showing **(A)** C1q deposition at the NMJ and **(B)** C1q expression by macrophages in support of the hypothesis of C1q tagging synapses for elimination.

Figure 3. Free Drug (anti-C1q) Detected in Plasma and Tissue Lysates Following Treatment

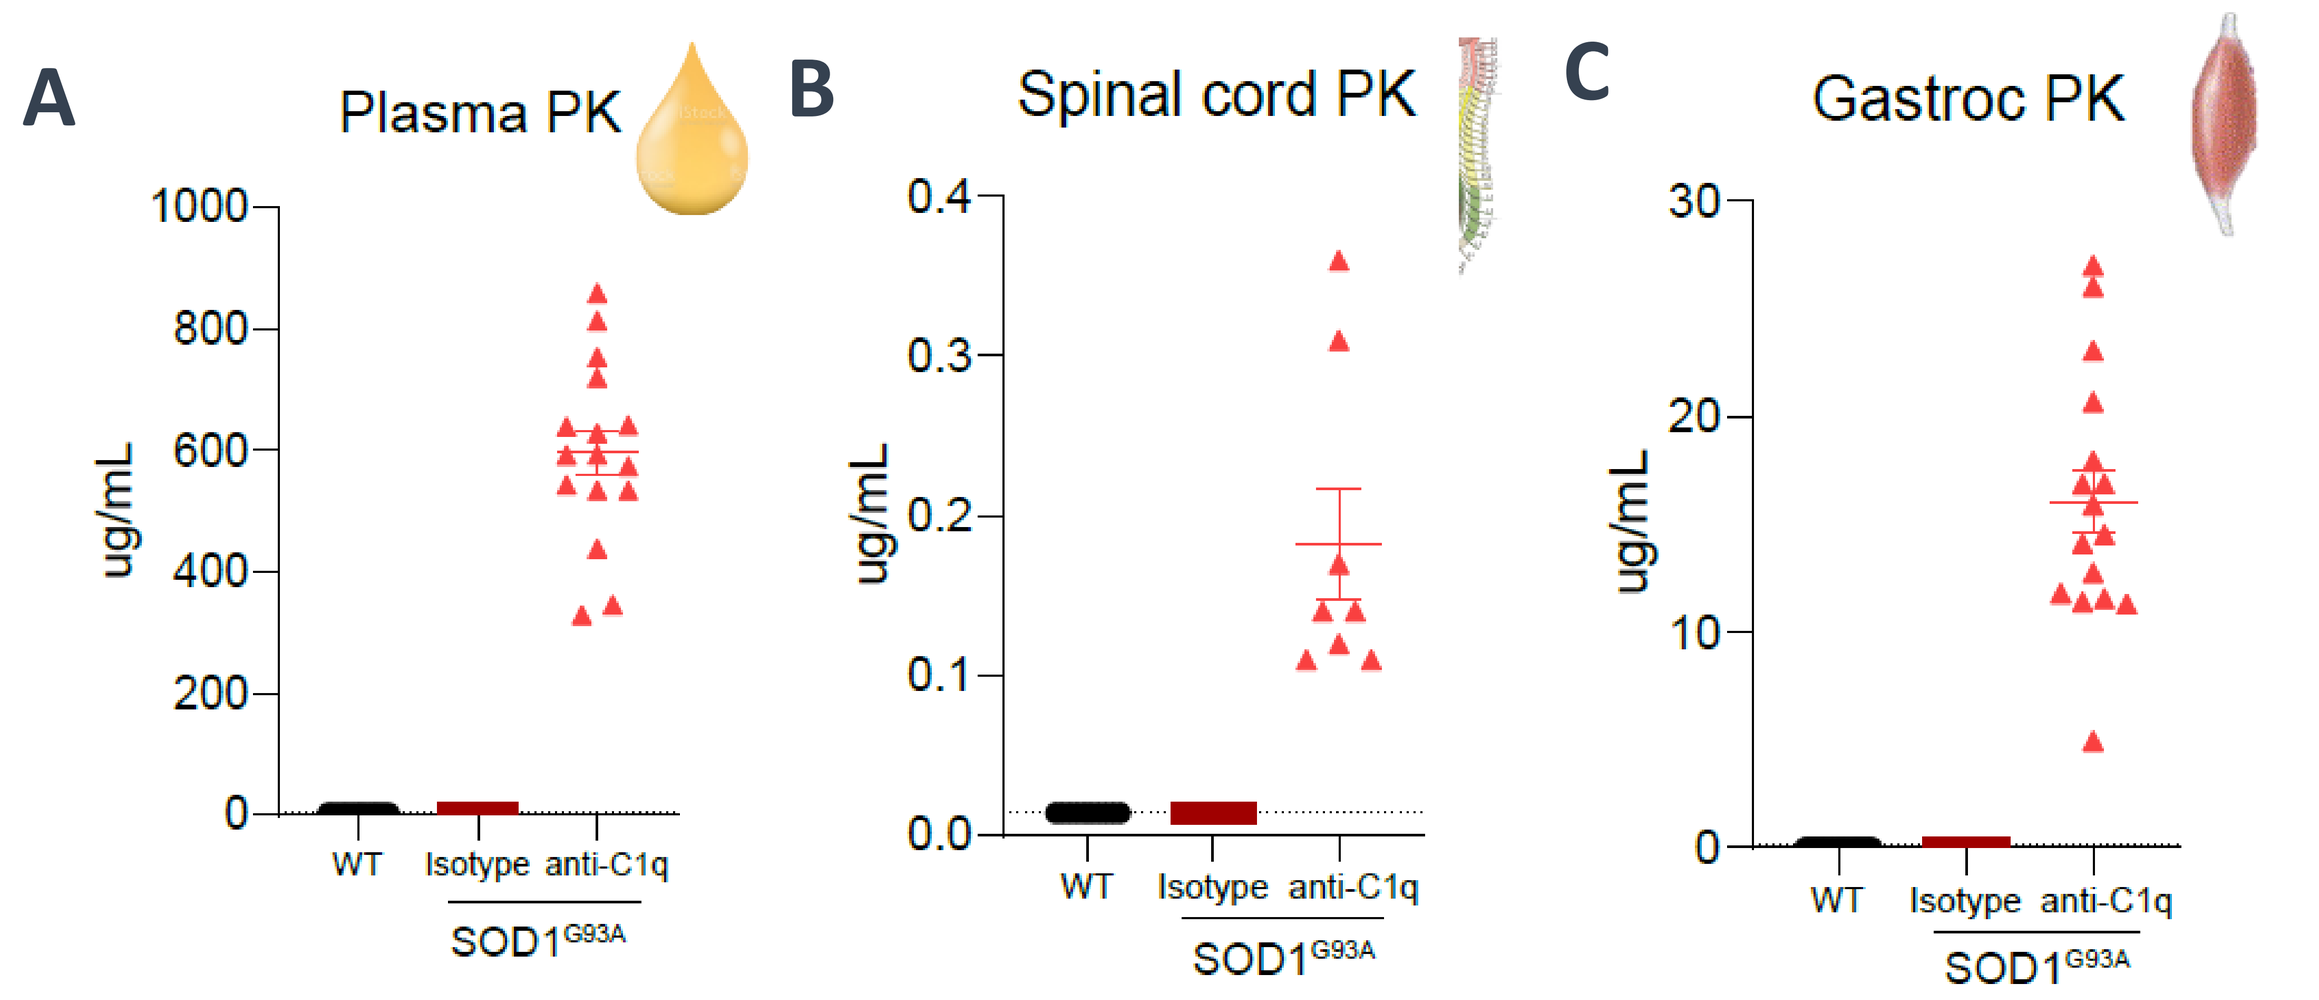

A
Plasma PK
B
Spinal cord PK
C
Gastroc PK
ug/mL
WT
Isotype
anti-C1q
SOD1G93A

Anti-C1q antibody levels were measured using standard ELISA in plasma, gastrocnemius and spinal cord tissue. Free antibody levels were measured in anti-C1q antibody treated SOD1^{G93A} mice, but not in WT or isotype treated SOD1^{G93A} mice.

Figure 4. C1q Target Engagement in Plasma, Spinal Cord, and Muscle Tissues

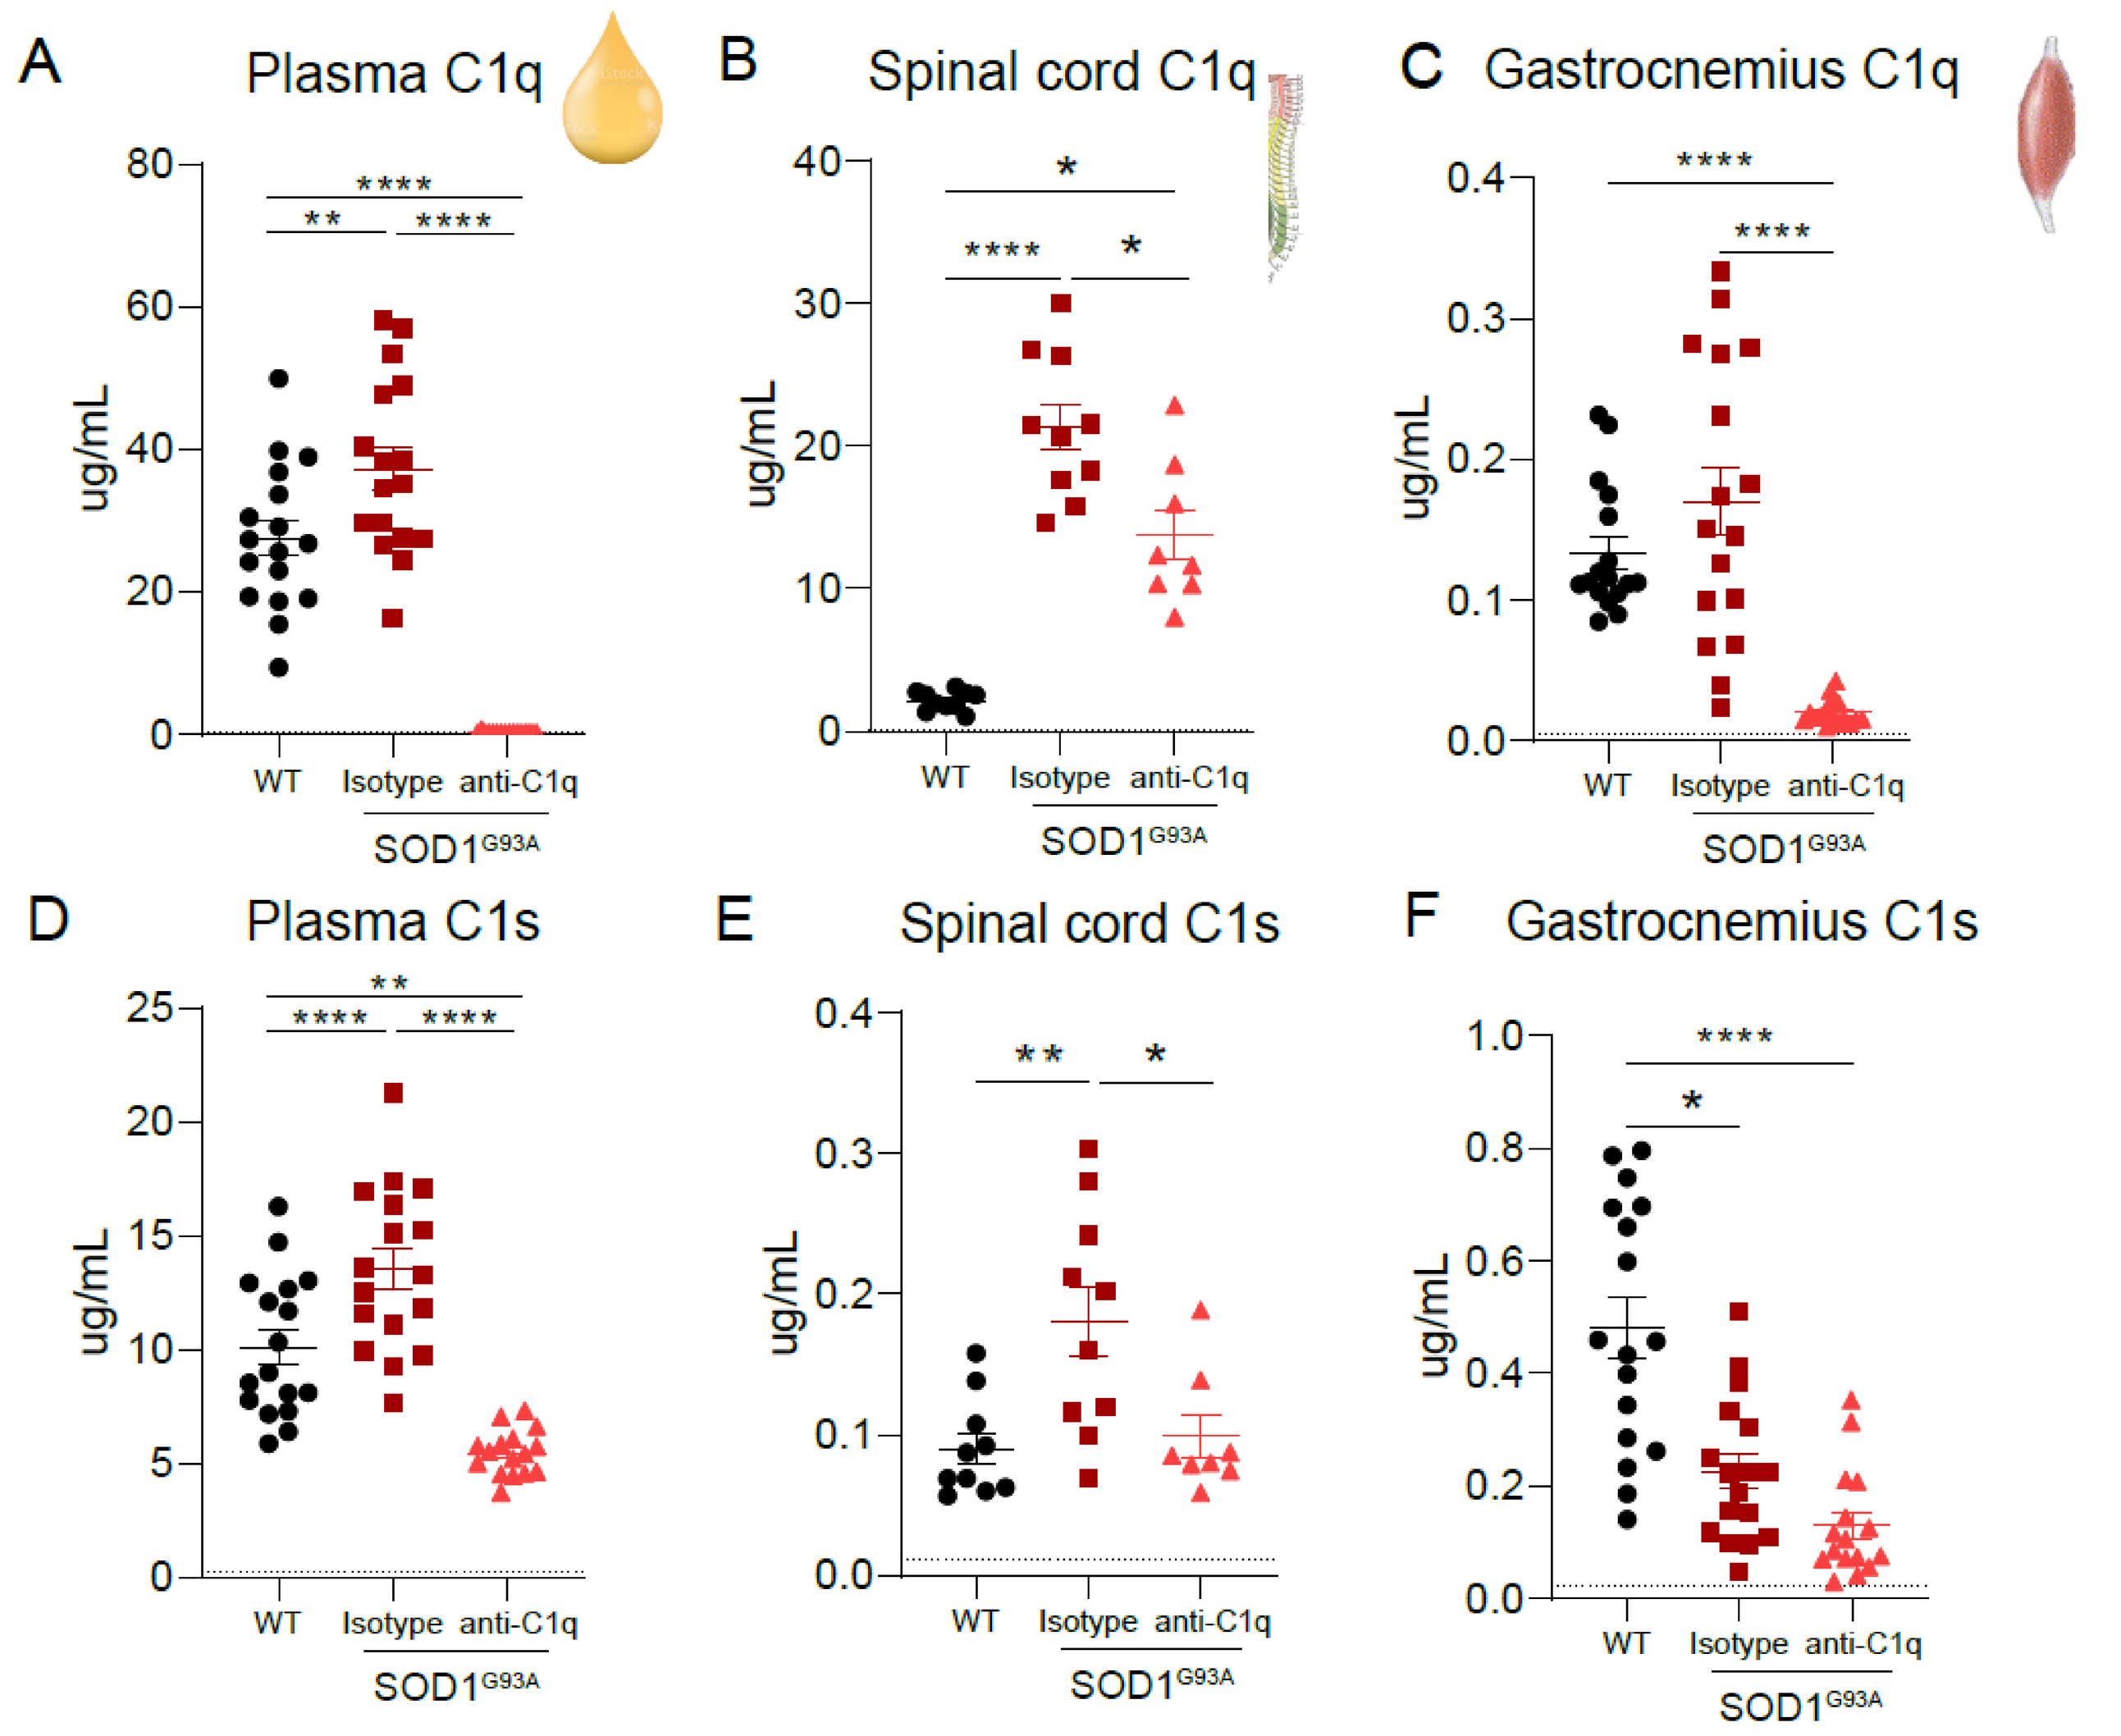

A
Plasma C1q
B
Spinal cord C1q
C
Gastrocnemius C1q
D
Plasma C1s
E
Spinal cord C1s
F
Gastrocnemius C1s
ug/mL
WT
Isotype
anti-C1q
SOD1G93A

Both C1q **(A-C)** and C1s **(D-F)** levels were increased in SOD1^{G93A} tissues compared to WT. Anti-C1q antibody treatment significantly reduced C1q as well as the downstream C1s in SOD1^{G93A} mice, indicating C1q engagement/inhibition within the spinal cord. The level of C1q engagement within the interstitial fluid of the spinal cord is likely to be higher than depicted since both intracellular and extracellular C1q will be exposed in tissue lysate. Statistical analysis: One-way ANOVA with repeated measures *P<0.05, **P<0.01, ****P<0.0001. Dotted line shows lower limit of quantification (C1q= 0.004-0.2 ug/mL, C1s= 0.02-0.2 ug/mL).

Figure 5. C1q Inhibition Reduces Neuronal Damage in SOD1^{G93A} Mice and CSF Nf-L Correlates with C1q Levels in Spinal Cord

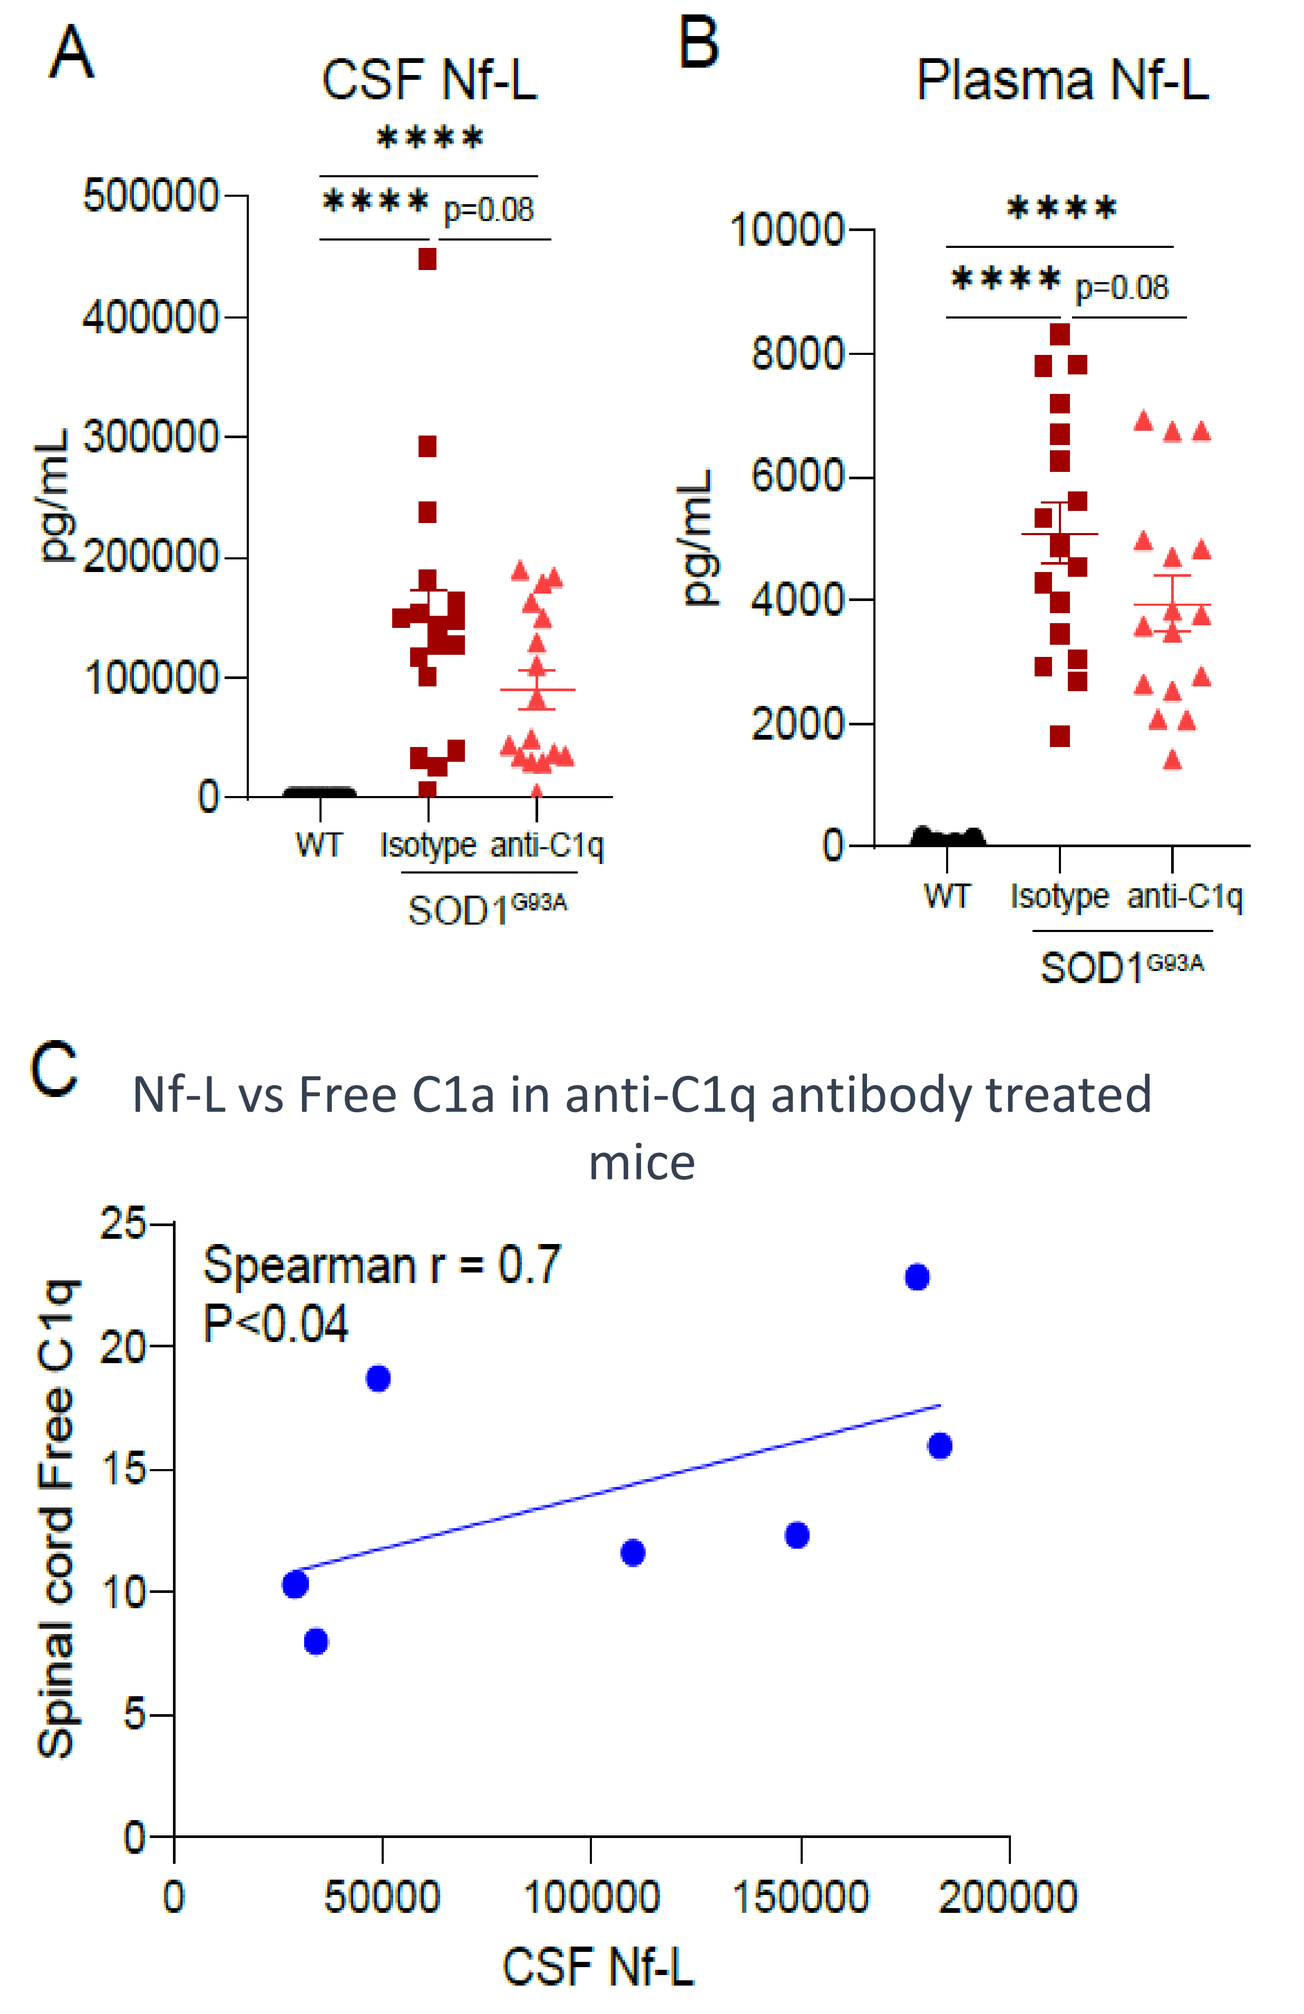

A
CSF Nf-L
B
Plasma Nf-L
pg/mL
p=0.08
WT
Isotype
anti-C1q
SOD1G93A
C
Nf-L vs Free C1a in anti-C1q antibody treated mice
Spearman r = 0.7
P<0.04
Spinal cord Free C1q
CSF Nf-L

Neurofilament-light chain (Nf-L) was measured as a marker of neuronal damage in mouse CSF and plasma after 9 weeks of anti-C1q antibody treatment.
SOD1^{G93A} mice showed ~600-fold and ~100-fold upregulation of Nf-L in CSF **(A)** and plasma **(B)** respectively. Anti-C1q antibody treatment reduced Nf-L in both CSF and plasma by 40%, suggesting that blocking the classical complement pathway reduces neuronal damage in SOD1^{G93A} mice. CSF Nf-L correlated significantly with reduced C1q levels in spinal cord of SOD1^{G93A} mice **(C)**. Statistical analysis: One-way ANOVA with repeated measures ****P<0.0001. Dotted line shows lower limit of quantification (1.52-7.6 pg/mL).

Figure 5. C1q Inhibition Improves CMAP in SOD1G93A Mice

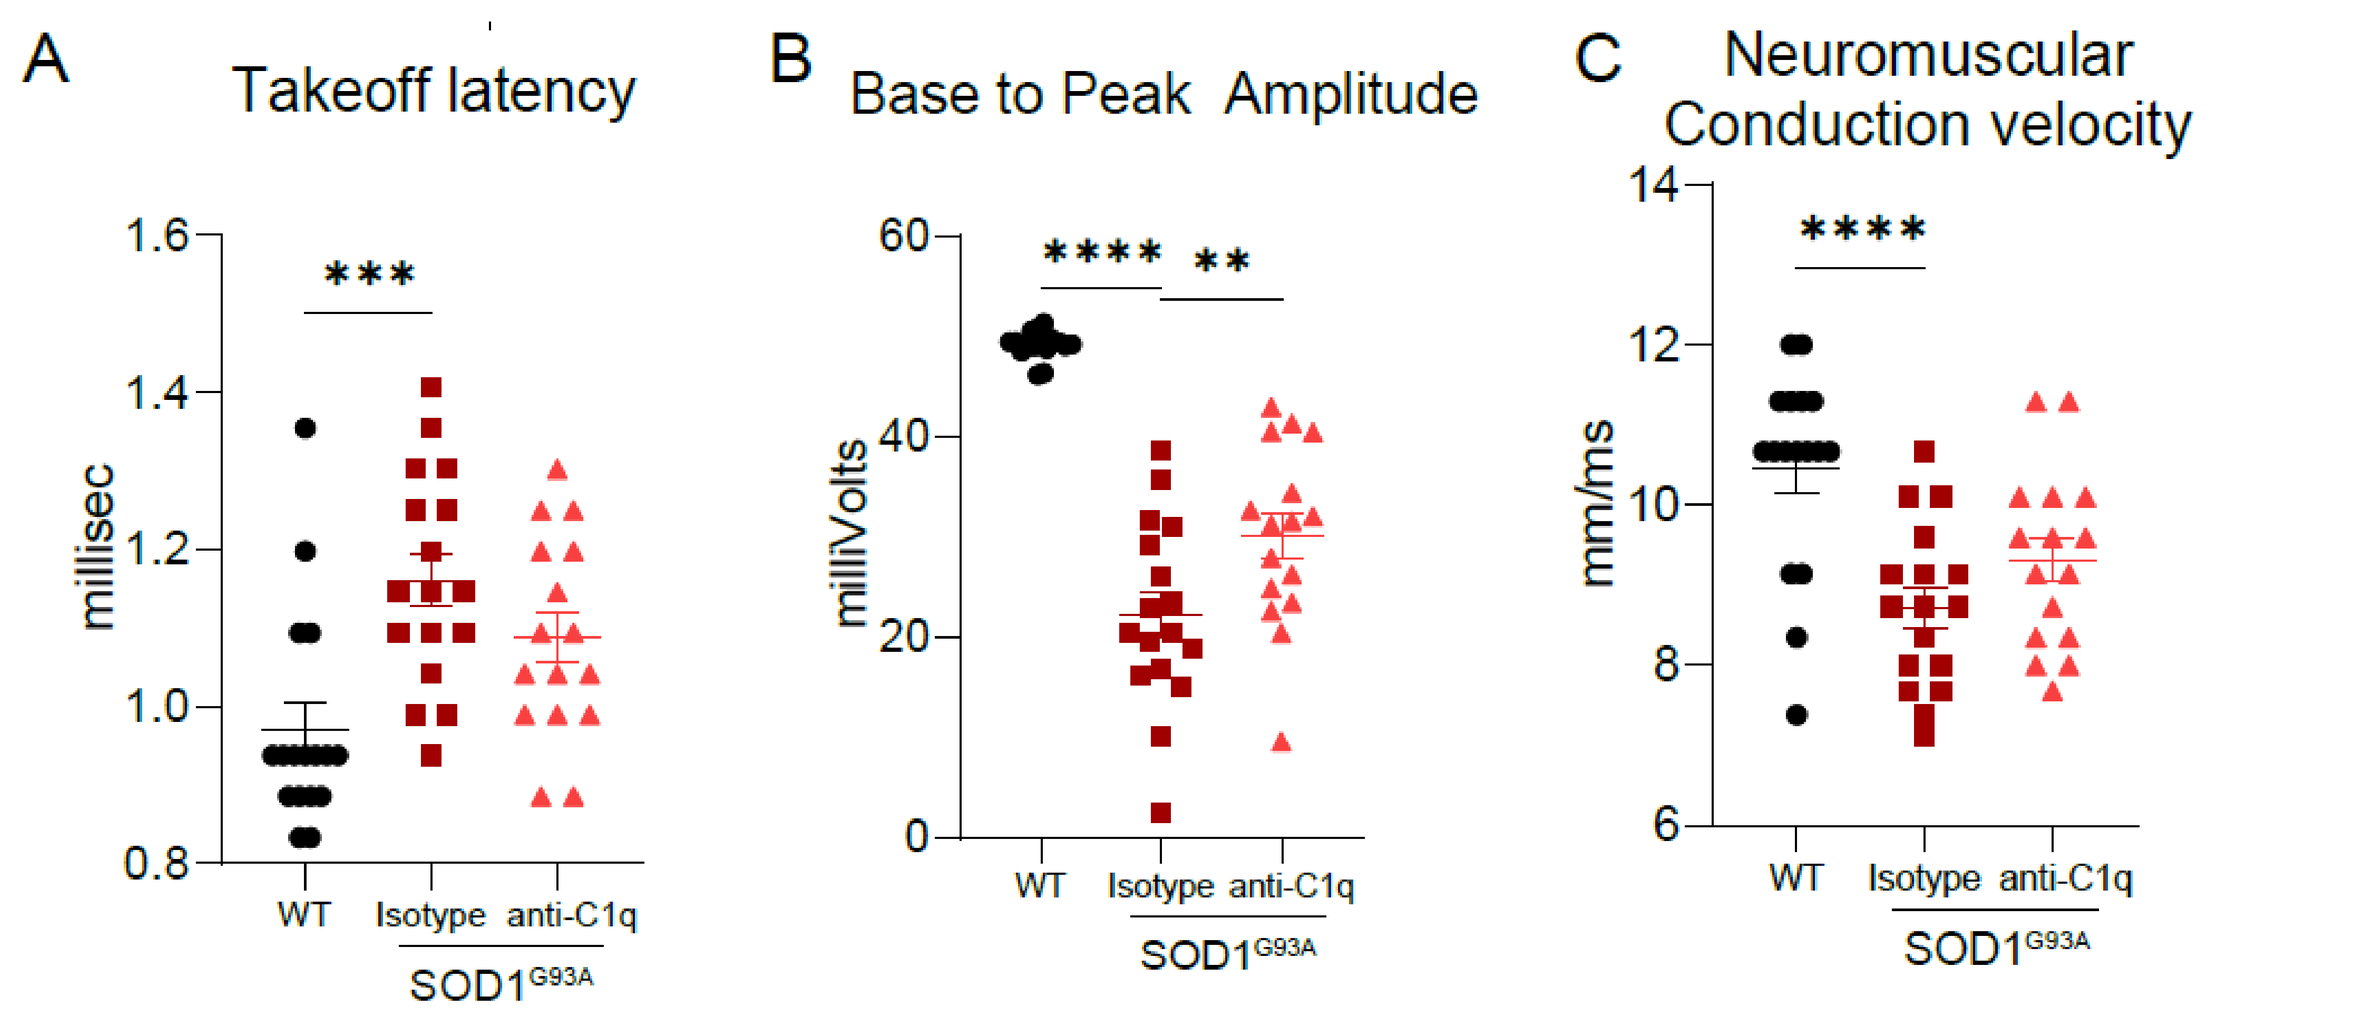

A
Takeoff latency
millisec
B
Base to Peak Amplitude
milliVolts
C
Neuromuscular Conduction velocity
mm/ms
WT
Isotype
anti-C1q
SOD1G93A

CMAP was measured to assess nerve conduction in the gastrocnemius muscle after 9 weeks of anti-C1q antibody treatment in SOD1^{G93A} mice. SOD1^{G93A} mice showed significantly increased latency (A), reduced amplitude (B) and reduced conduction velocity (C). Anti-C1q antibody treatment significantly improved CMAP amplitude (B) in SOD1^{G93A} mice which marks improvement in axonal preservation and/or synaptic connections at the NMJ. Statistical analysis: One-way ANOVA with repeated measures **P<0.01, ***P<0.001, ****P<0.0001.

Figure 6. Anti-C1q Preserves NMJ in SOD1 Mouse Model

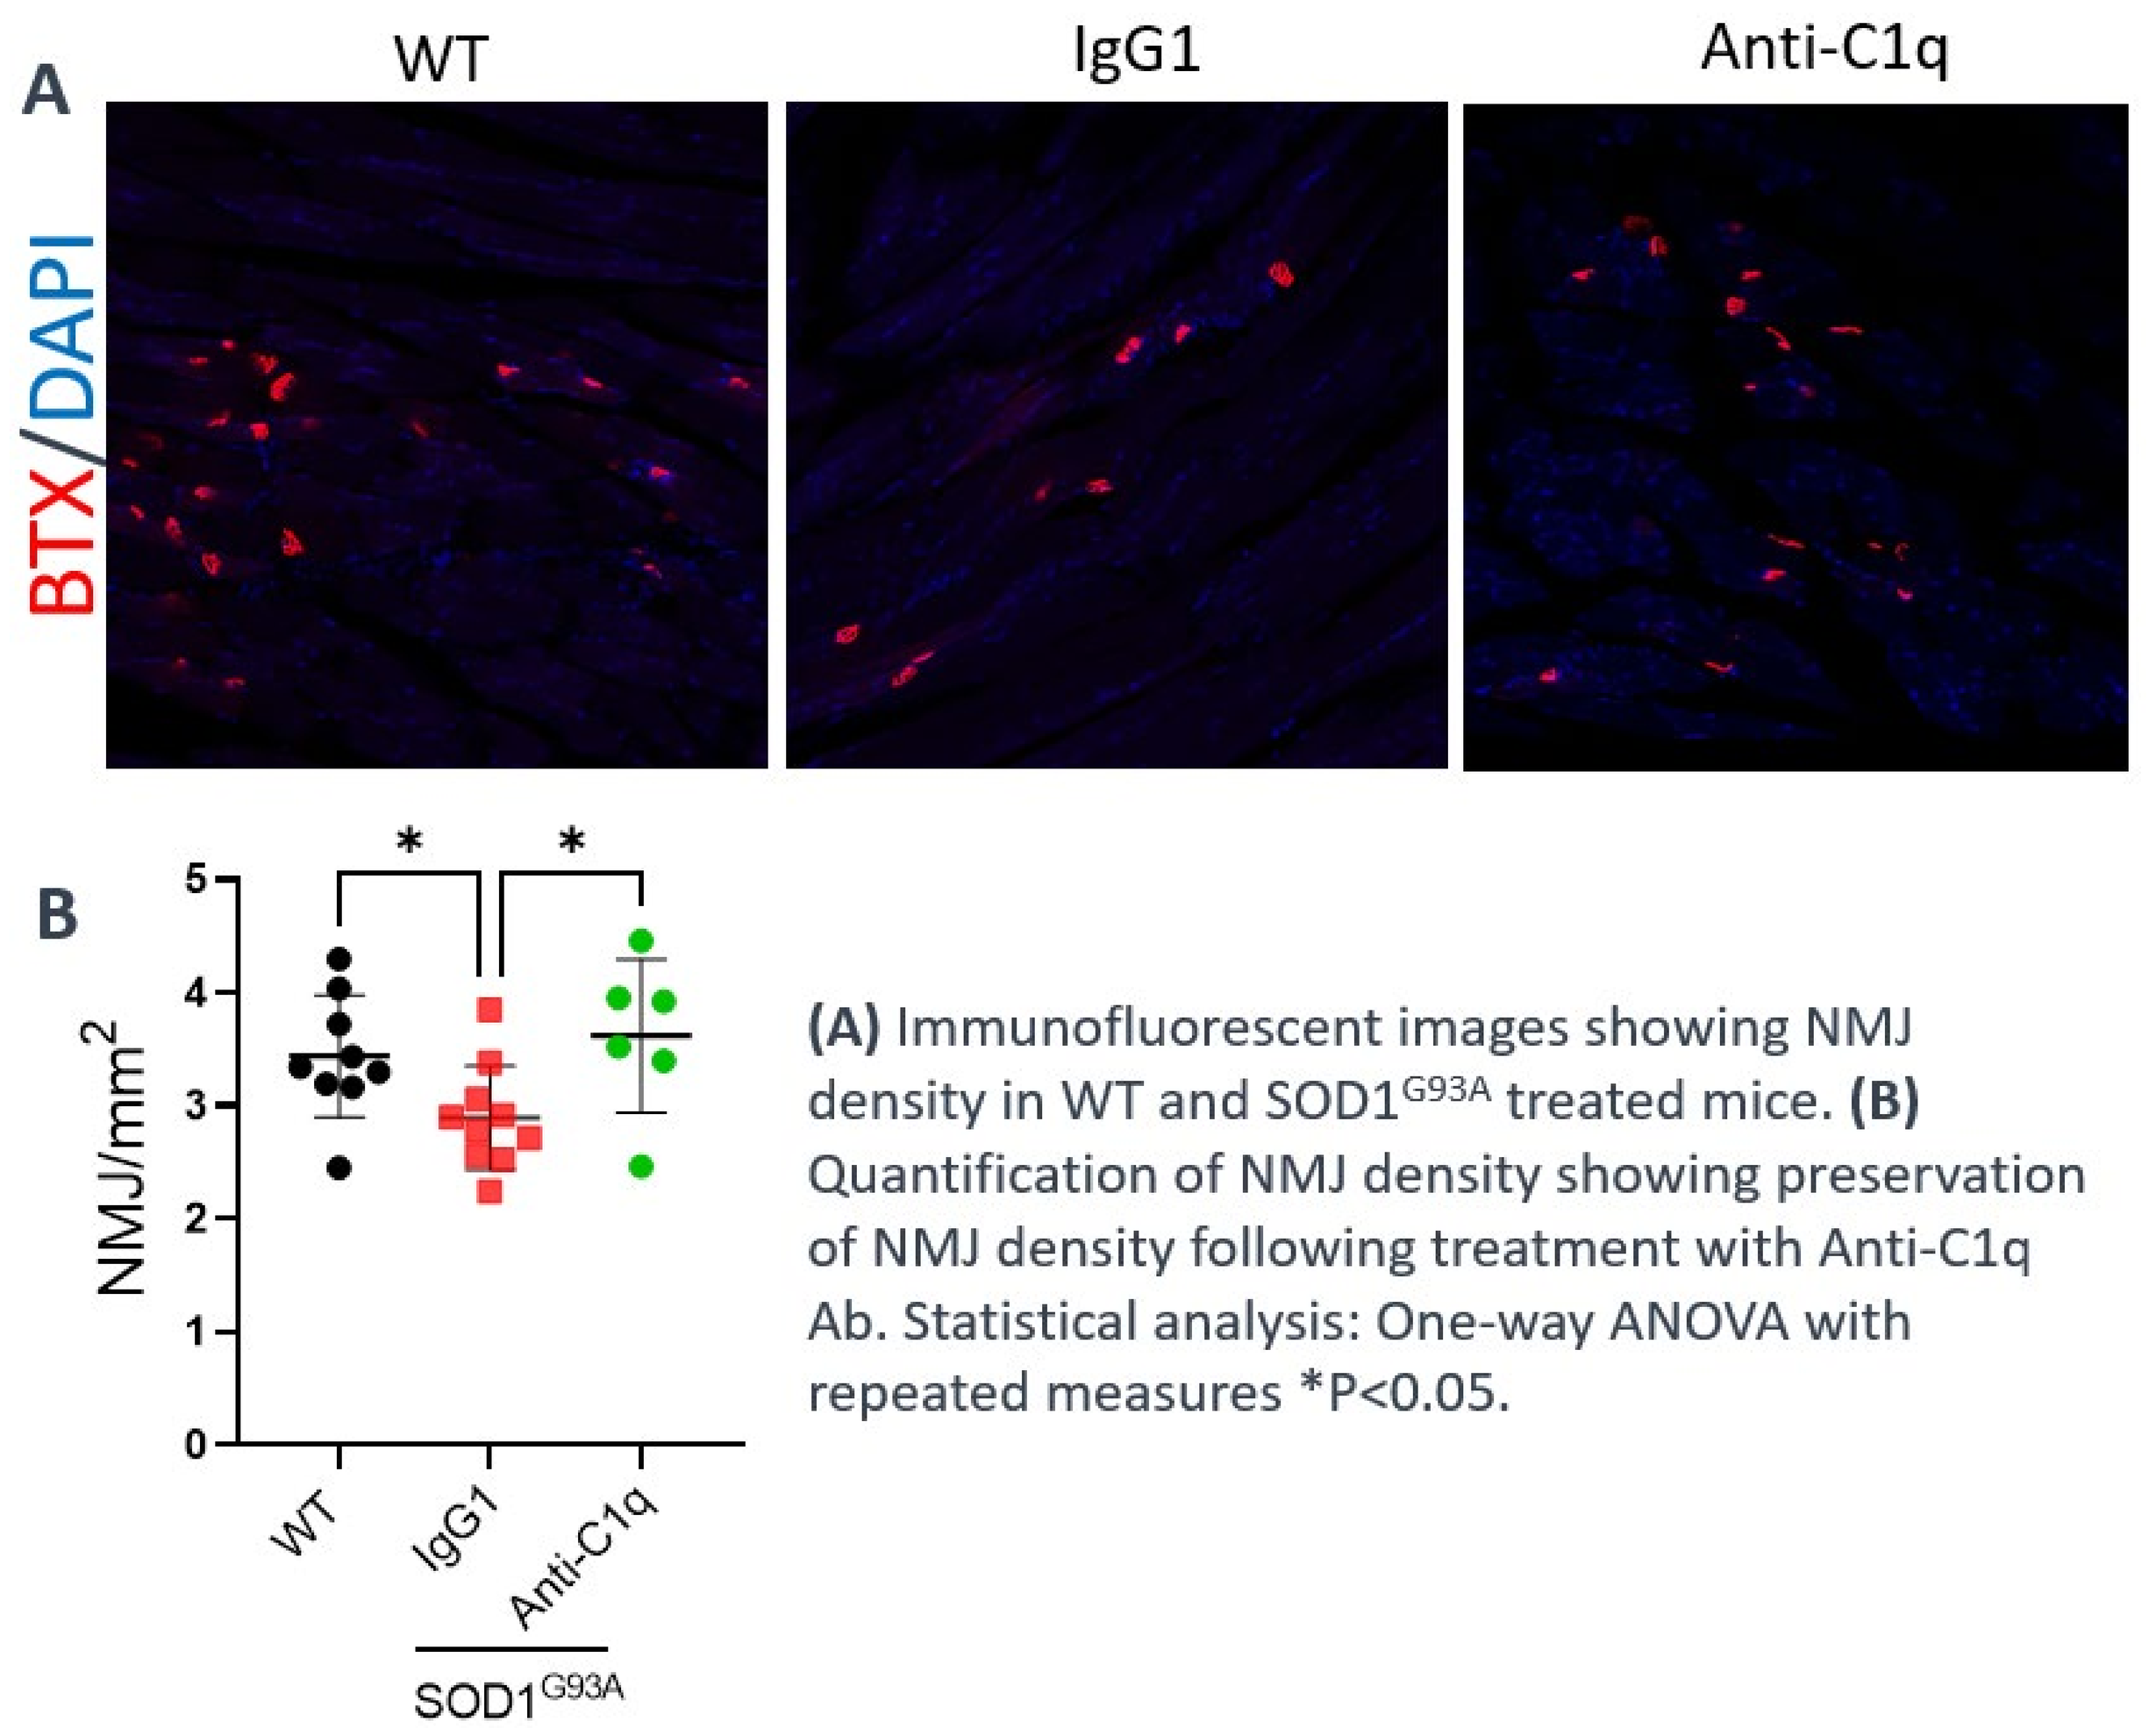

A
WT
IgG1
Anti-C1q
BTX/DAPI
B
NMJ/mm2
WT
IgG1
Anti-C1q
SOD1G93A

(A) Immunofluorescent images showing NMJ density in WT and SOD1^{G93A} treated mice. **(B)** Quantification of NMJ density showing preservation of NMJ density following treatment with Anti-C1q Ab. Statistical analysis: One-way ANOVA with repeated measures *P<0.05.

Conclusions

- We provide evidence of C1q deposition at the NMJ and expression by infiltrating macrophages in the SOD1^{G93A} mouse model of ALS
- We show that inhibition of classical complement pathway results in reduced neuronal damage, as measured by Nf-L, improved muscle nerve conduction and NMJ preservation in the SOD1^{G93A} mice
- Classical complement component C1q represents a potential pharmacological target in ALS. A Phase 2 study of ANX005, an anti-C1q therapy, in ALS patients is ongoing (clinicaltrials.gov NCT04569435).

Society for Neuroscience

#453.13