C1q Inhibition Protects Photoreceptor Synapses and Preserves Retinal Function in a Preclinical Model of Photoreceptor Degeneration

A0463

Alessia Tassoni, Christina Huynh, Joseph Vereen, Serafin Gatiga, Yaisa Andrews-Zwilling, Donald Fong, Daniel Emerling, Rick Artis, Ted Yednock

Annexon Biosciences, 1400 Sierra Point Pkwy, 2nd Floor, Brisbane, CA 94080

BACKGROUND

Geographic atrophy (GA) is an advanced form of age-related macular degeneration leading to photoreceptor death and visual loss. C1q-activating substrates and increased expression of complement components have been observed in retinal tissue from GA patients, yet the role of complement in disease progression and visual loss is unknown. In a recent phase 2 clinical trial (clinicaltrials.gov NCT04656561), we reported that intravitreal administration of anti-C1q (ANX007) significantly preserved visual function in patients with GA over a 12-month treatment period. However, the mechanism of action (MOA) through which C1q pharmacological inhibition provides visual function protection has not been investigated. We hypothesize that C1q tags photoreceptor synapses of the outer plexiform layer (OPL), driving synapse pruning. Blocking C1q and classical cascade activation will prevent synapse loss, promote neuronal survival and preserve retinal function.

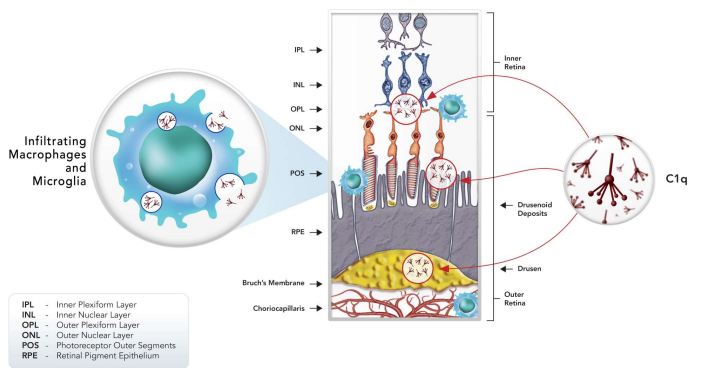

Infiltrating Macrophages and Microglia
IPL
INL
OPL
ONL
POS
RPE
Bruch's Membrane
Choriocapillaris
Inner Retina
Drusenoid Deposits
Drusen
Outer Retina
C1q
IPL - Inner Plexiform Layer
INL - Inner Nuclear Layer
OPL - Outer Plexiform Layer
ONL - Outer Nuclear Layer
POS - Photoreceptor Outer Segments
RPE - Retinal Pigment Epithelium

OBJECTIVES

- To determine C1q and classical complement pathway involvement in synapse loss in a model of photoreceptor cell degeneration
- To determine the therapeutic benefit of classical complement inhibition in protecting photoreceptor synapses and preserving retinal function

METHOD

Light damage: BALB/c mice were exposed to white light to cause retinal damage and observed at Days 1, 3 and 7 post-light exposure (acute: 25k lux for 4 h; mild: 5k lux for 30 min).
Complement signature: Classical complement component levels were measured in retinal lysates by standard sandwich ELISA.
C1q deposition on synapses and microglial engulfment: C1q expression in the tissue was assessed by immunofluorescence (IF) and confocal microscopy. Microglial engulfment of synapses was assessed using IMARIS software.
C1q inhibition: C1q activity was pharmacologically blocked by intraperitoneal injection of a C1q inhibitory antibody one day prior to light exposure. Tissue was assessed at Day 5 after treatment by ELISA and IF.
Human tissue procurement: Retina specimens from GA patients were procured from the San Diego Eye Bank.

RESULTS

Figure 1. Progressive Photoreceptor Synapse Loss and Increased Microgliosis in the Light Damage Model

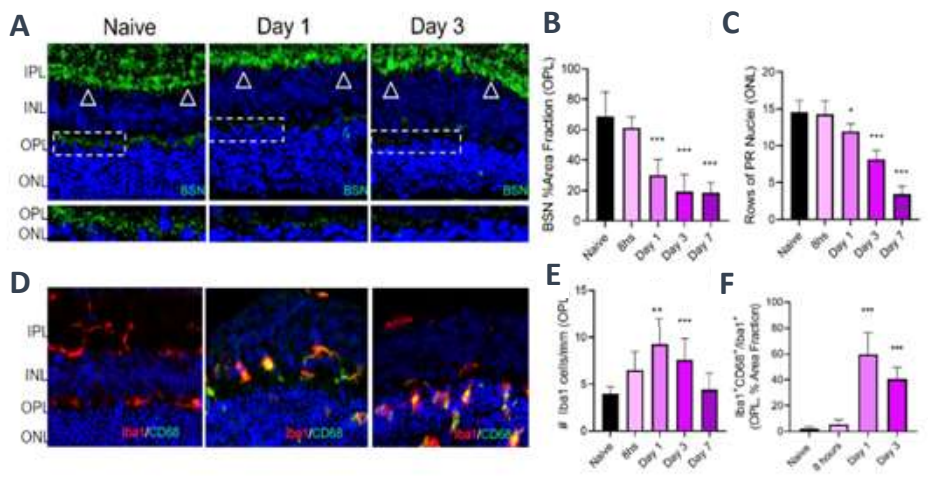

A
Naive
Day 1
Day 3
IPL
INL
OPL
ONL
BSN
B
BSN %Area Fraction (OPL)
C
Rows of PR Nuclei (ONL)
Naive
8hs
Day 1
Day 3
Day 7
D
Iba1/CD68
E
Iba1 cells/mm (OPL)
F
Iba1+CD68+/Iba1+ (OPL, % Area Fraction)

(A-C) Progressive loss of photoreceptor synapses (Bassoon, green) and cell bodies (DAPI, blue) following light damage assessed by IF. (D-F) Increased microglia/macrophage reactivity (Iba1, red and CD68, yellow) following light damage assessed by IF. Distribution of phagocytic microglia in the synaptic layer peaked at Day 1, when significant synapse loss was first observed. *P<0.05, **P<0.01, ***P<0.001.

Figure 2. Increased C1q Expression and Deposition Within the OPL Correlates With Photoreceptor Synapse Loss in the Light Damage Model

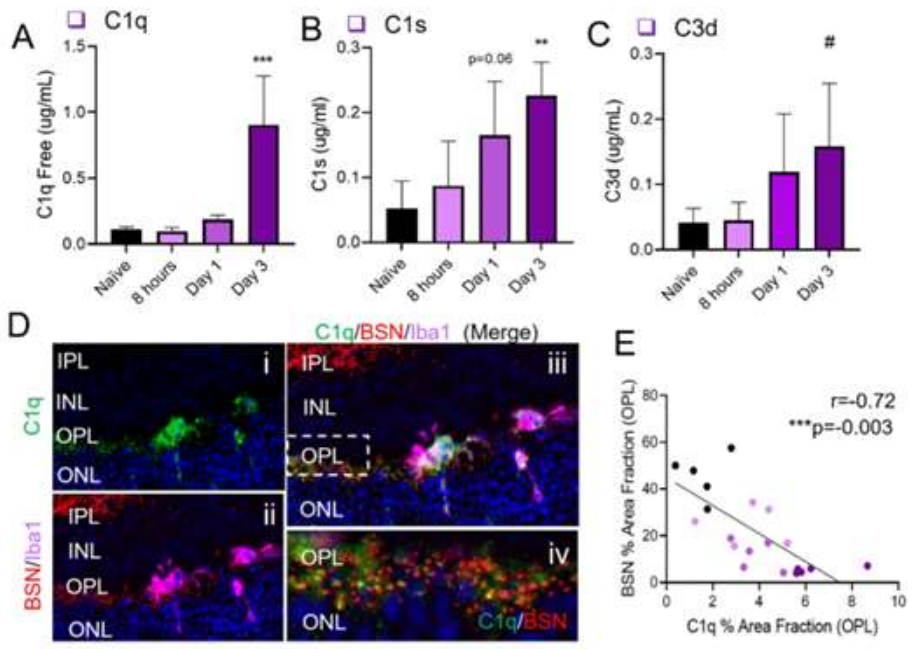

A
C1q
C1q Free (ug/mL)
B
C1s
C1s (ug/ml)
p=0.06
C
C3d
C3d (ug/mL)
#
Naive
8 hours
Day 1
Day 3
D
C1q/BSN/Iba1 (Merge)
C1q
BSN/Iba1
IPL
INL
OPL
ONL
C1q/BSN
E
BSN % Area Fraction (OPL)
C1q % Area Fraction (OPL)
r=-0.72
***p=-0.003

(A-C) Increased levels of the initiating classical complement components C1q and C1s, as well as the downstream activation product C3d, in retinal lysate following light damage assessed by ELISA. (D) IF showing retinal C1q distribution (green) and its colocalization with microglia/macrophage (Iba1, magenta) and synapses (Bassoon, red). (E) Correlation analysis showing significant negative correlation between C1q levels in the OPL and photoreceptor synaptic density, consistent with a causal relationship. *P<0.05, **P<0.01, ***P<0.001.

Figure 3. Microglial Cell Engulfment of C1q-Tagged Photoreceptor Synapses Following Light Damage

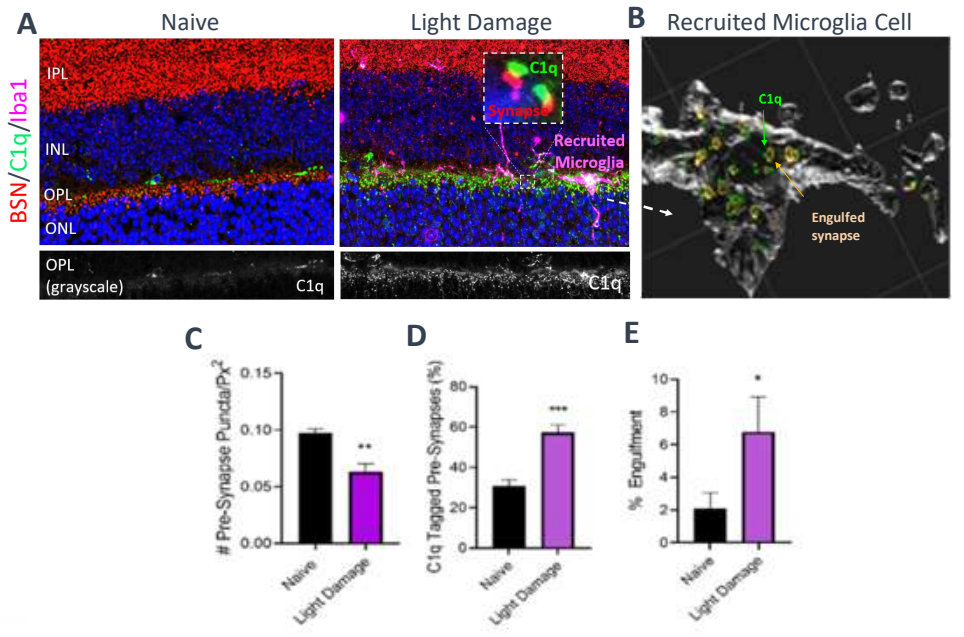

A
Naive
Light Damage
BSN/C1q/Iba1
IPL
INL
OPL
ONL
C1q
Synapse
Recruited Microglia
OPL (grayscale)
B
Recruited Microglia Cell
Engulfed synapse
C
Pre-Synapse Puncta/Px²
D
C1q Tagged Pre-Synapses (%)
E
% Engulfment
Naive
Light Damage

(A) Increased C1q levels (green) in the OPL of light-damaged retina and its proximity to synapses (red) observed by IF. (B) Microglial engulfment of C1q-tagged synapses in light-damaged retina demonstrated by high-resolution and 3D surface rendering. (C-E) Quantitative analysis showing significant decrease in synapse density (C), increase in the percentage of C1q-tagged synapses (D), and increase in microglial engulfed C1q-tagged synapses (E) in light-damaged retina. *P<0.05, **P<0.01, ***P<0.001.

Figure 4. Phosphatidylserine (PS) Externalization and C1q Deposition on Photoreceptor Synapses *In Vivo* After Light Damage and *In Vitro* Binding of C1q to PS

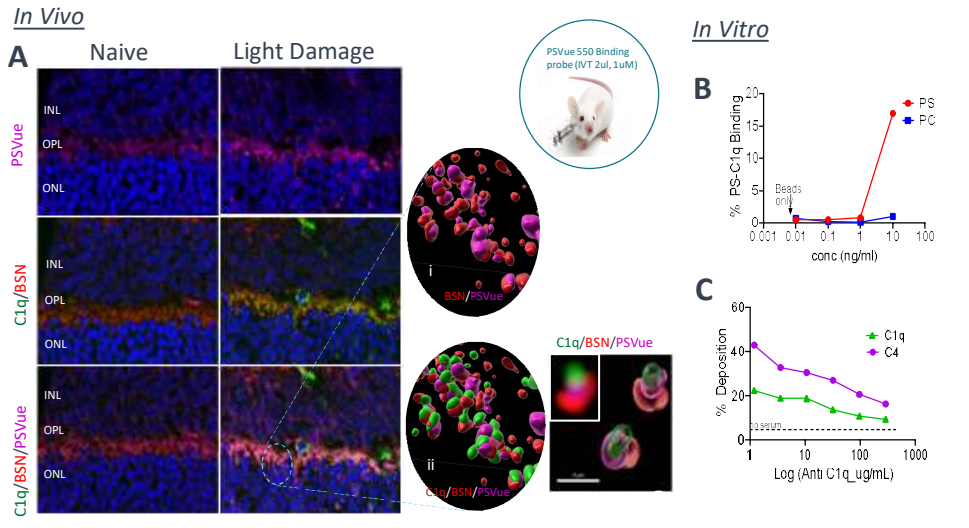

In Vivo
A
Naive
Light Damage
PSVue
C1q/BSN
C1q/BSN/PSVue
INL
OPL
ONL
BSN/PSVue
PSVue 550 Binding probe (IVT 2ul, 1uM)
In Vitro
B
% PS-C1q Binding
Beads only
conc (ng/ml)
PS
PC
C
% Deposition
C1q
C4
Log (Anti C1q_ug/mL)

(A) PS externalization on synapses demonstrated by PSVue labelling of PS (magenta) in the OPL of light-damaged retina. 3D surface rendered images (i-ii) showing PSVue proximity to Bassoon and C1q (magenta, red and green). (B) C1q to PS binding confirmed by in vitro binding assay using PS-coated lipid beads; no binding to control phosphatidylcholine (PC) beads. (C) In vitro competition binding assay demonstrating anti-C1q antibody efficacy in reducing deposition of C1q and C4 on serum exposed PS lipid beads.

Figure 5. Anti-C1q Treatment Reduces Synaptic Loss and Preserves Retinal Function in the Light Damage Model

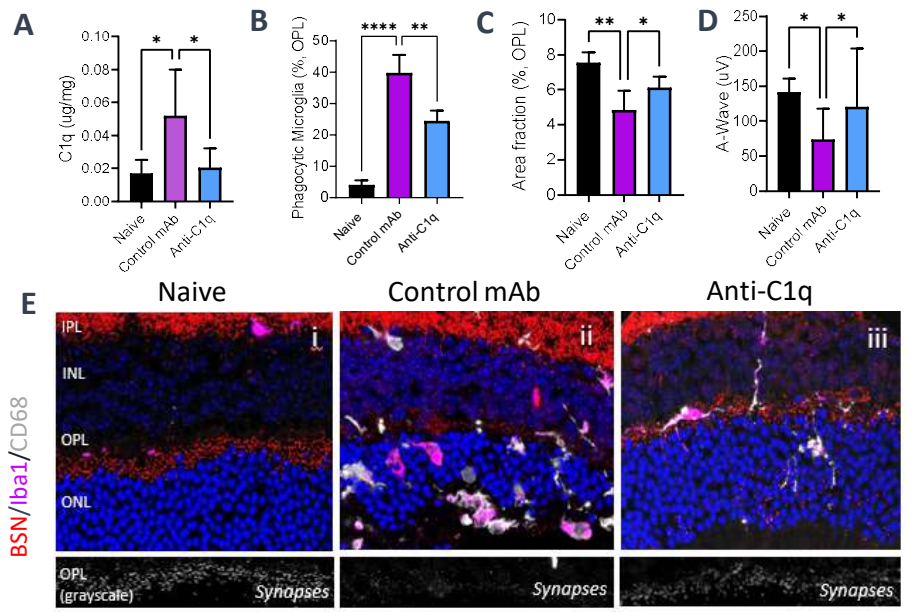

A
C1q (ug/mg)
B
Phagocytic Microglia (%, OPL)
C
Area fraction (%, OPL)
D
A-Wave (uV)
Naive
Control mAb
Anti-C1q
E
BSN/Iba1/CD68
IPL
INL
OPL
ONL
OPL (grayscale)
Synapses

(A) Target engagement confirmed by ELISA showing C1q decrease in retina upon anti-C1q treatment. (B-D) Reduced inflammation, preserved photoreceptor synapses, and maintained retinal function following anti-C1q treatment. (E) Photoreceptor synaptic protection (red) and reduced microglial reactivity (Iba, magenta and CD68, grey) observed by IF in retina following anti-C1q treatment. *P<0.05, **P<0.01, ***P<0.001.

Figure 6. C1q Deposition on Photoreceptor Synapses and Their Loss Outside Lesion Edge in Human GA Postmortem Retina

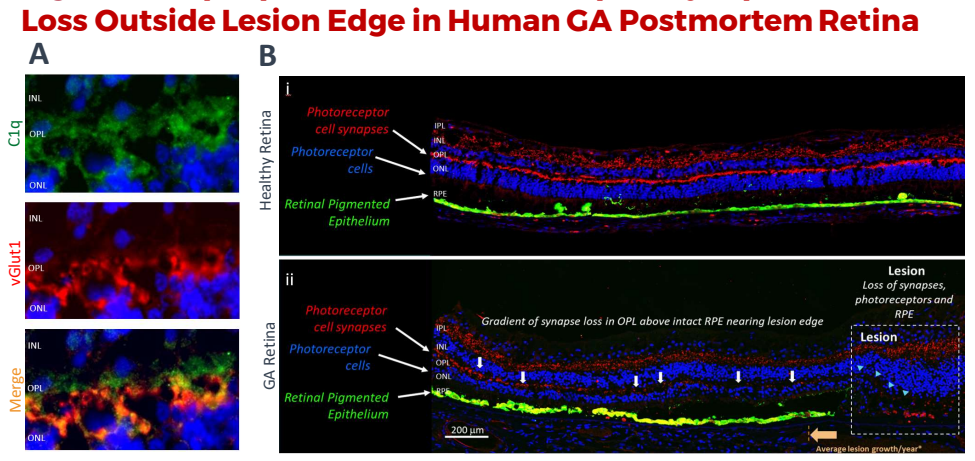

A
C1q
vGlut1
Merge
INL
OPL
ONL
B
Healthy Retina
GA Retina
Photoreceptor cell synapses
Photoreceptor cells
Retinal Pigmented Epithelium
Lesion
Loss of synapses, photoreceptors and RPE
Gradient of synapse loss in OPL above intact RPE nearing lesion edge
200 µm
Average lesion growth/year*

(A) C1q deposition (green) on photoreceptor synapses (red) in OPL of human GA postmortem retina observed by IF. (B) Loss of photoreceptor synapses (red) in GA retina (ii) compared to healthy retina (i) demonstrated by IF. In GA retina, synapse loss is observed outside area of atrophy, defined by retinal pigment epithelium layer integrity (green). These data demonstrate that neurodegeneration occurs well outside atrophic areas in the GA retina.

CONCLUSIONS

This study demonstrates C1q pathogenic role in photoreceptor synapse elimination in neurodegeneration and provides mechanistic understanding of anti-C1q role in visual function preservation in GA, reported in our phase 2 clinical trial (NCT04656561).

DISCLOSURES

AT, CH, JV, YA, DRA, and TY: employment with an equity ownership in Annexon Biosciences, Inc.; SG, DF, and DE: employment with Annexon Biosciences, Inc.