EPO-0633

Tanruprubart Rapidly Improves Health-Related Quality of Life in Patients With Guillain-Barré Syndrome: A Randomised, Placebo-Controlled Phase 3 Study

Myoung Kim[1], Glenn Morrison[1], Jing Du[1], Quazi Deen Mohammad[2], Nowshin Papri[3], Henk-André Kroon[1], Kenneth C Gorson[4]

[1]Annexon Biosciences, Brisbane, CA, USA; [2]National Institute of Neuroscience (NINS), Dhaka, Bangladesh; [3]icddr,b, Dhaka, Bangladesh; [4]Tufts University School of Medicine, Boston, MA, USA

Scan to download

Introduction

- Guillain-Barré syndrome (GBS) is a rare, rapidly progressive neuromuscular emergency that can affect anyone at anytime, often requiring prolonged hospitalisation and intensive care, and in some cases can be fatal[1]
- GBS is often associated with life-long disability.[1-3] Patients with long-term residual effects often need to change their work and general lifestyle to adapt after acute recovery[4]
- Tanruprubart (ANX005), a monoclonal antibody, is a targeted immunotherapy that selectively binds to and inhibits C1q, the initiating molecule of the classical complement pathway, thus providing rapid inhibition of complement-mediated neuroinflammation and nerve damage[5,6]
- In GBS-02 (NCT04701164), a Phase 3, multicentre, double-blind, placebo-controlled trial of tanruprubart in patients with GBS, patients treated with tanruprubart 30 mg/kg had better outcomes (adjusted odds ratio [aOR] 2.4; 95% CI 1.3–4.5; p=0.0058) according to the GBS-Disability Score (GBS-DS) relative to placebo at Week 8[7]
- Tanruprubart was well tolerated, and most adverse events were mild to moderate in severity, attributed to GBS and not considered related to the study drug[7]
- This analysis presents patient-reported outcomes with tanruprubart 30 mg/kg vs placebo

Methods

GBS-02 study design

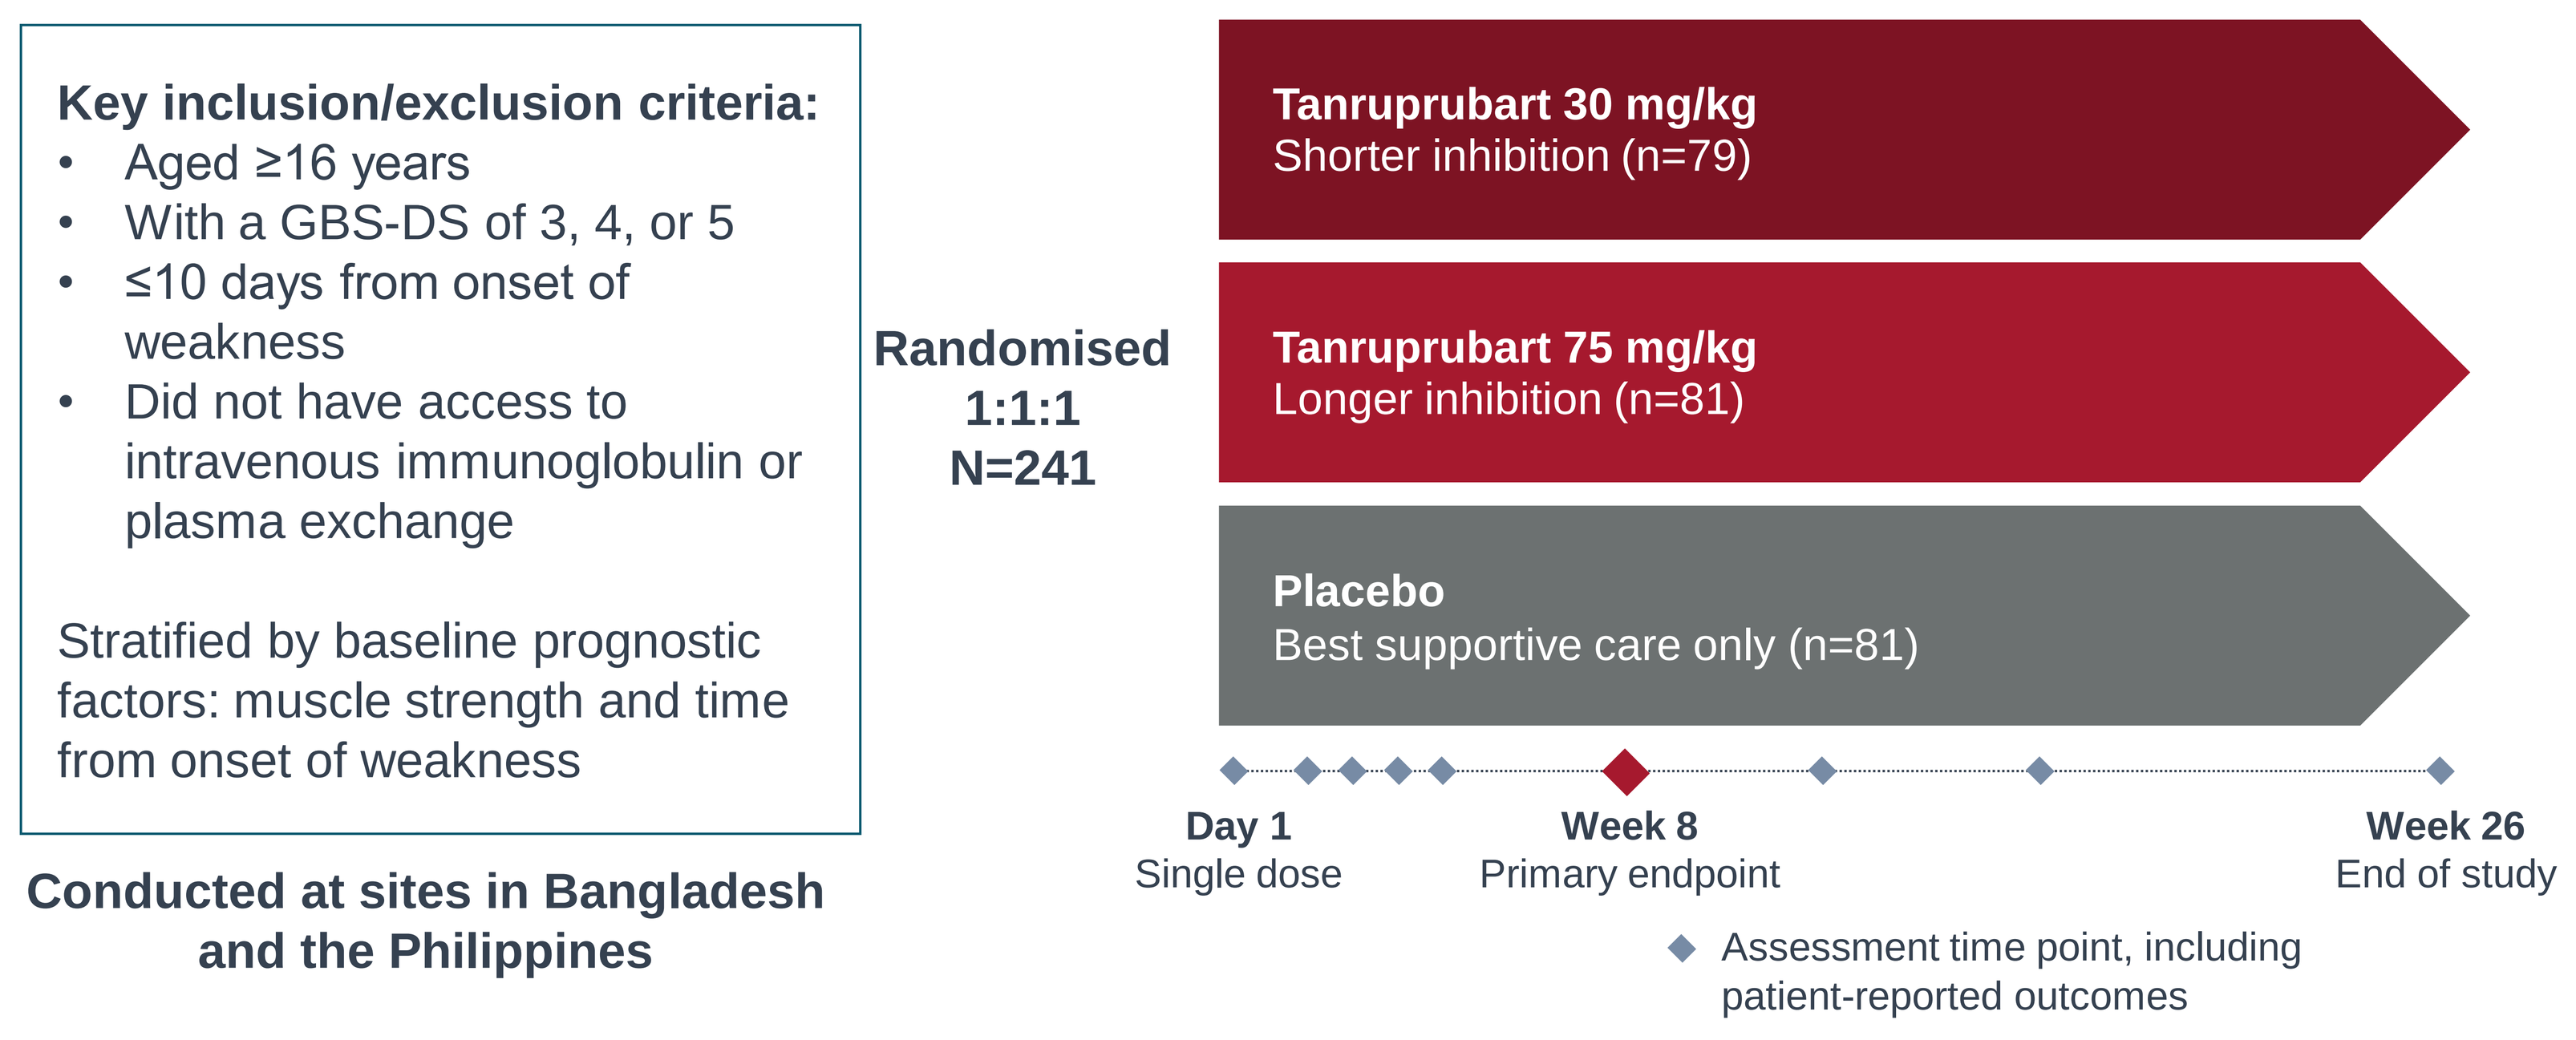

Key inclusion/exclusion criteria:
• Aged ≥16 years
• With a GBS-DS of 3, 4, or 5
• ≤10 days from onset of weakness
• Did not have access to intravenous immunoglobulin or plasma exchange
Stratified by baseline prognostic factors: muscle strength and time from onset of weakness
Randomised 1:1:1 N=241
Tanruprubart 30 mg/kg Shorter inhibition (n=79)
Tanruprubart 75 mg/kg Longer inhibition (n=81)
Placebo Best supportive care only (n=81)
Day 1 Single dose
Week 8 Primary endpoint
Week 26 End of study
Assessment time point, including patient-reported outcomes

Conducted at sites in Bangladesh and the Philippines

Patient reported outcomes:

- EuroQol 5-Dimension 5-Level tool (EQ-5D-5L)
- Rasch-built Modified Fatigue Severity Scale (rFSS)
- Patient Global Impression of Change (PGIC)
- Impact of treatment on health-related quality of life was evaluated using proportional odds logistic regression adjusted for baseline covariates

Results

- **EQ-5D-5L:** By Day 8, significant improvements occurred in mobility, self-care and usual activities with tanruprubart 30 mg/kg vs placebo which continued to Week 8. Improvements in pain/discomfort and anxiety/depression numerically favoured tanruprubart (**Figure 1**)
- **rFSS:** Improvements in all domains favoured tanruprubart compared with placebo at Day 8 and Week 8 except for "exercise brings on my fatigue" (**Figure 2**)
- **PGIC:** By Day 8, 8 patients (10.1%) reported being "very much improved" with tanruprubart vs 3 patients (3.7%) with placebo (aOR 2.5; 95% CI: 1.4–4.6; p=0.0028). Improvements at Week 8 still favoured tanruprubart (aOR 1.4; 95% CI 0.8–2.6, p=0.2917; **Figure 3**)

Figure 1. Patients reporting no problems (0) across EQ-5D-5L domains at Day 8 and Week 8

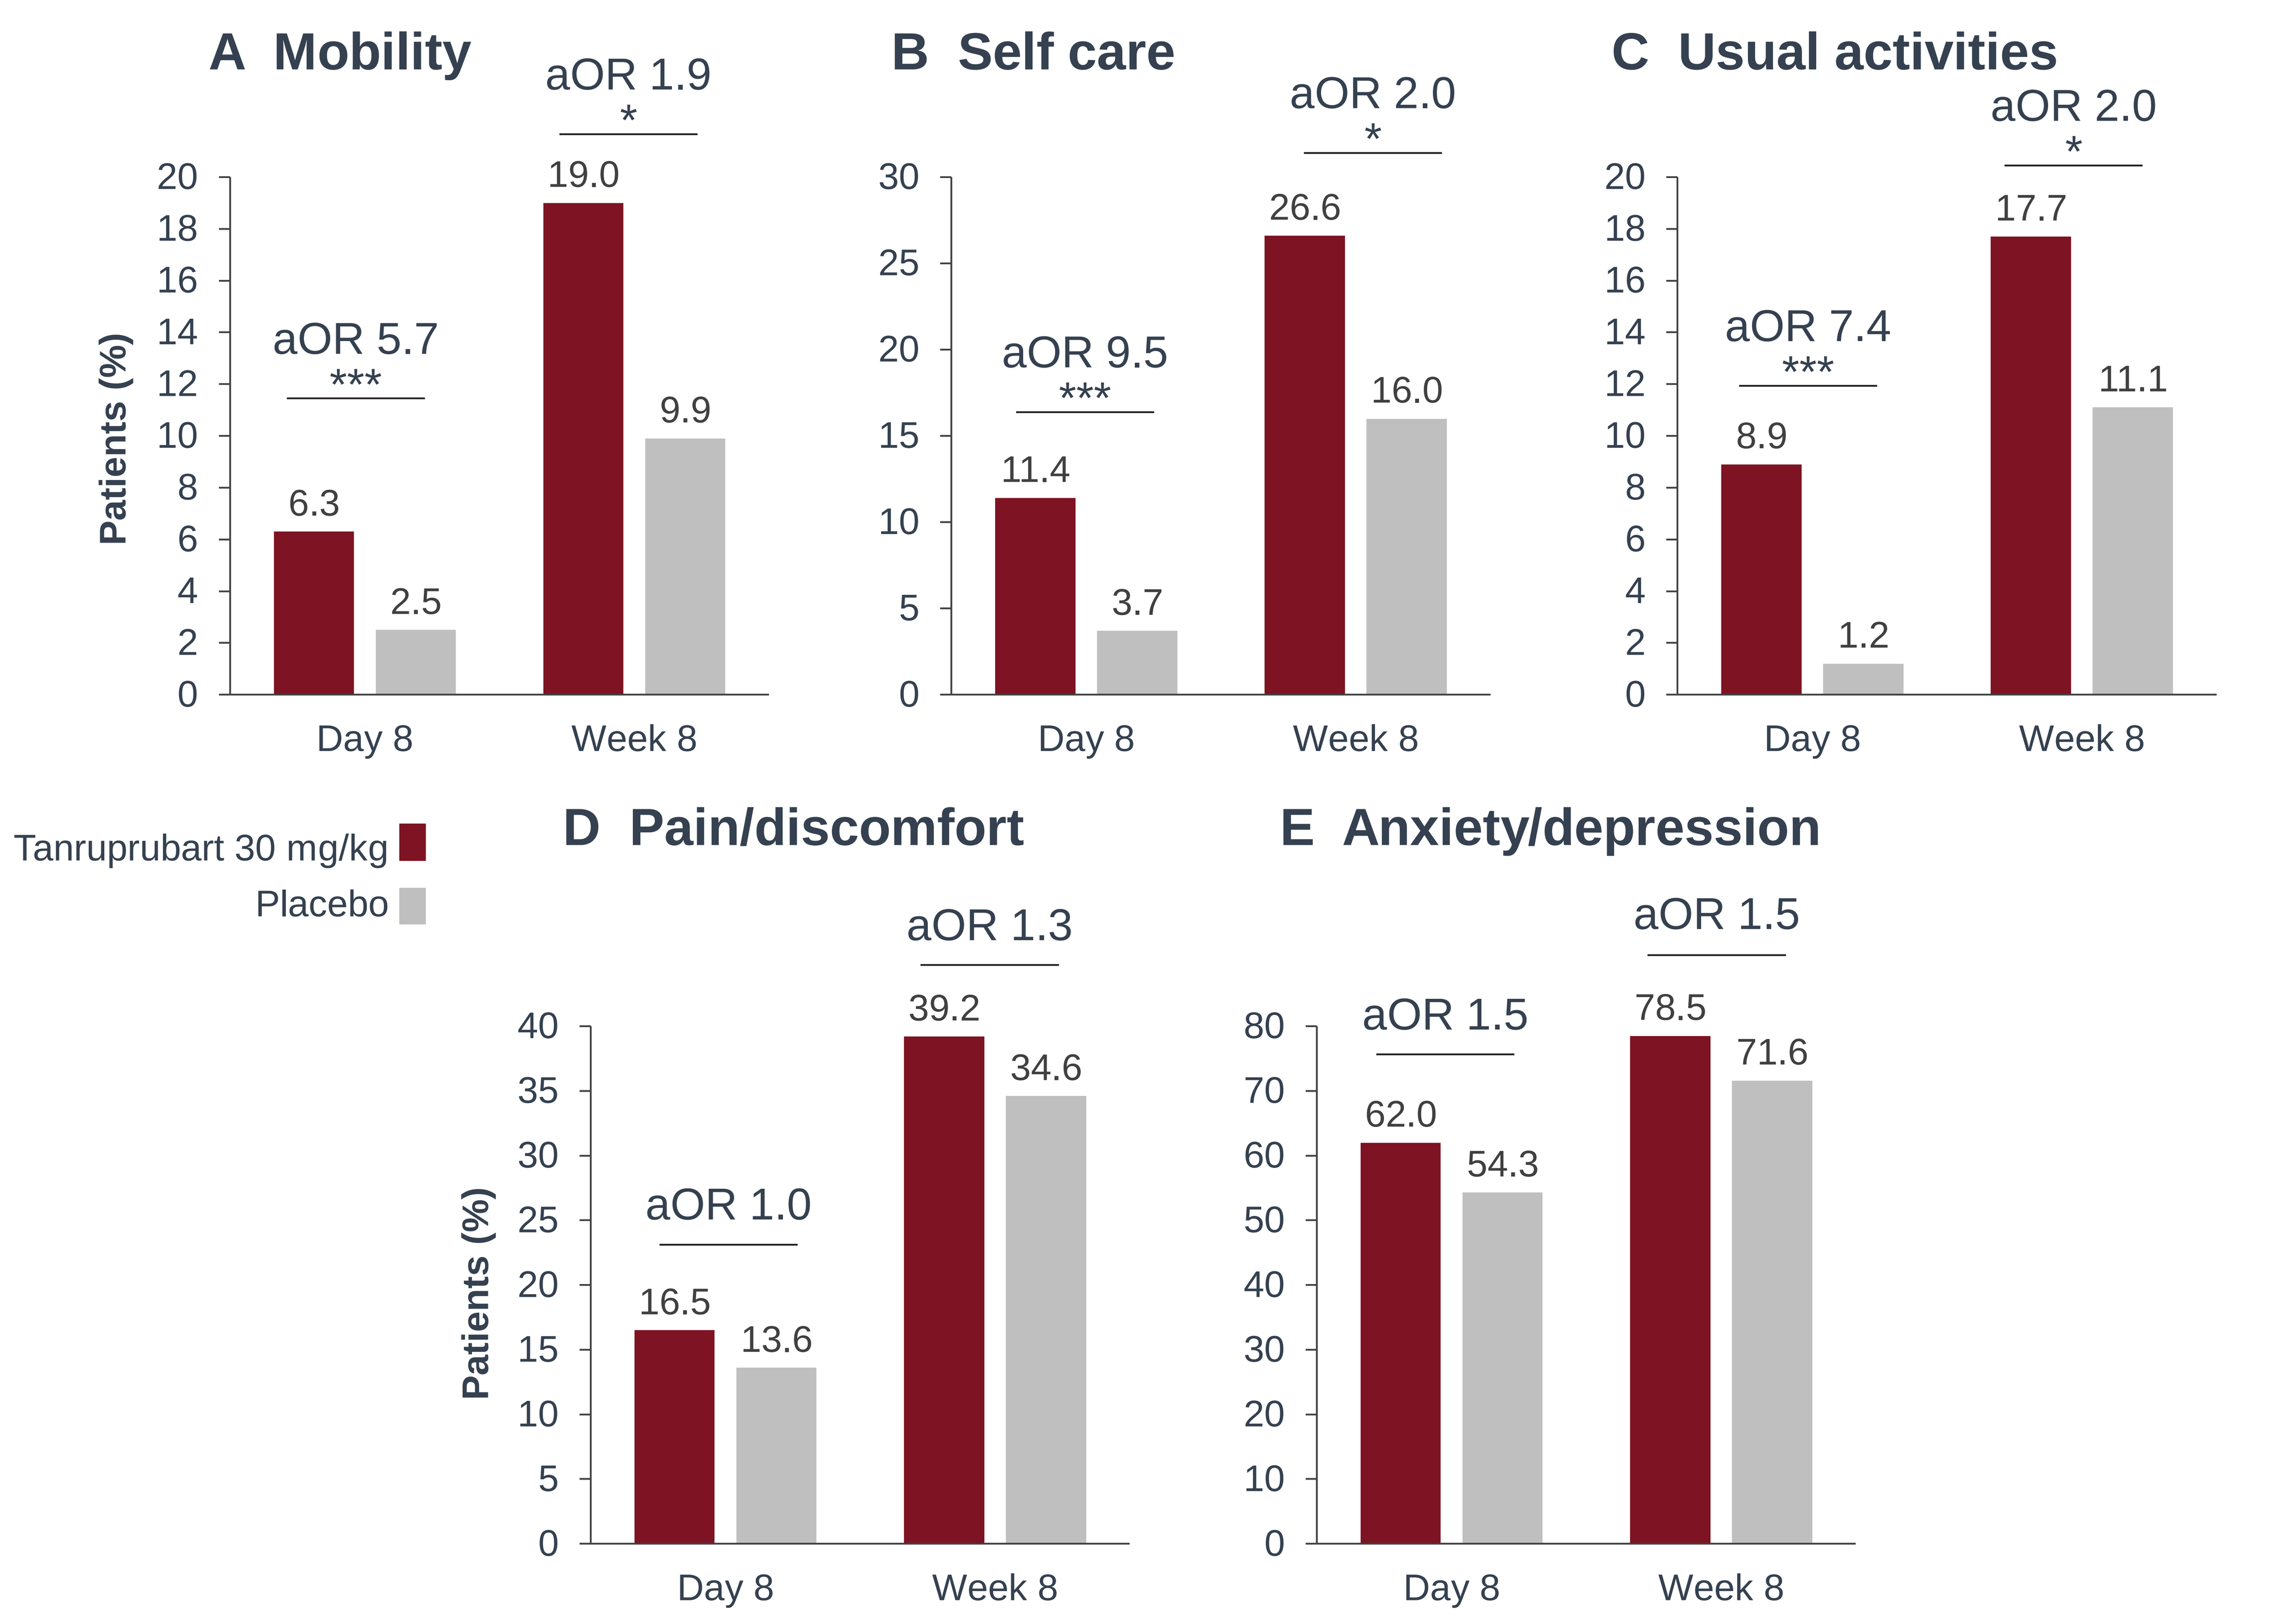

A Mobility
aOR 5.7 ***
aOR 1.9 *
6.3
2.5
19.0
9.9
B Self care
aOR 9.5 ***
aOR 2.0 *
11.4
3.7
26.6
16.0
C Usual activities
aOR 7.4 ***
aOR 2.0 *
8.9
1.2
17.7
11.1
Tanruprubart 30 mg/kg
Placebo
D Pain/discomfort
aOR 1.0
aOR 1.3
16.5
13.6
39.2
34.6
E Anxiety/depression
aOR 1.5
aOR 1.5
62.0
54.3
78.5
71.6
Patients (%)
Day 8
Week 8

***p<0.0001; **p<0.001; *p<0.05.

Figure 2. Patients who answered "disagree" to statements across rFSS domains at Day 8 and Week 8

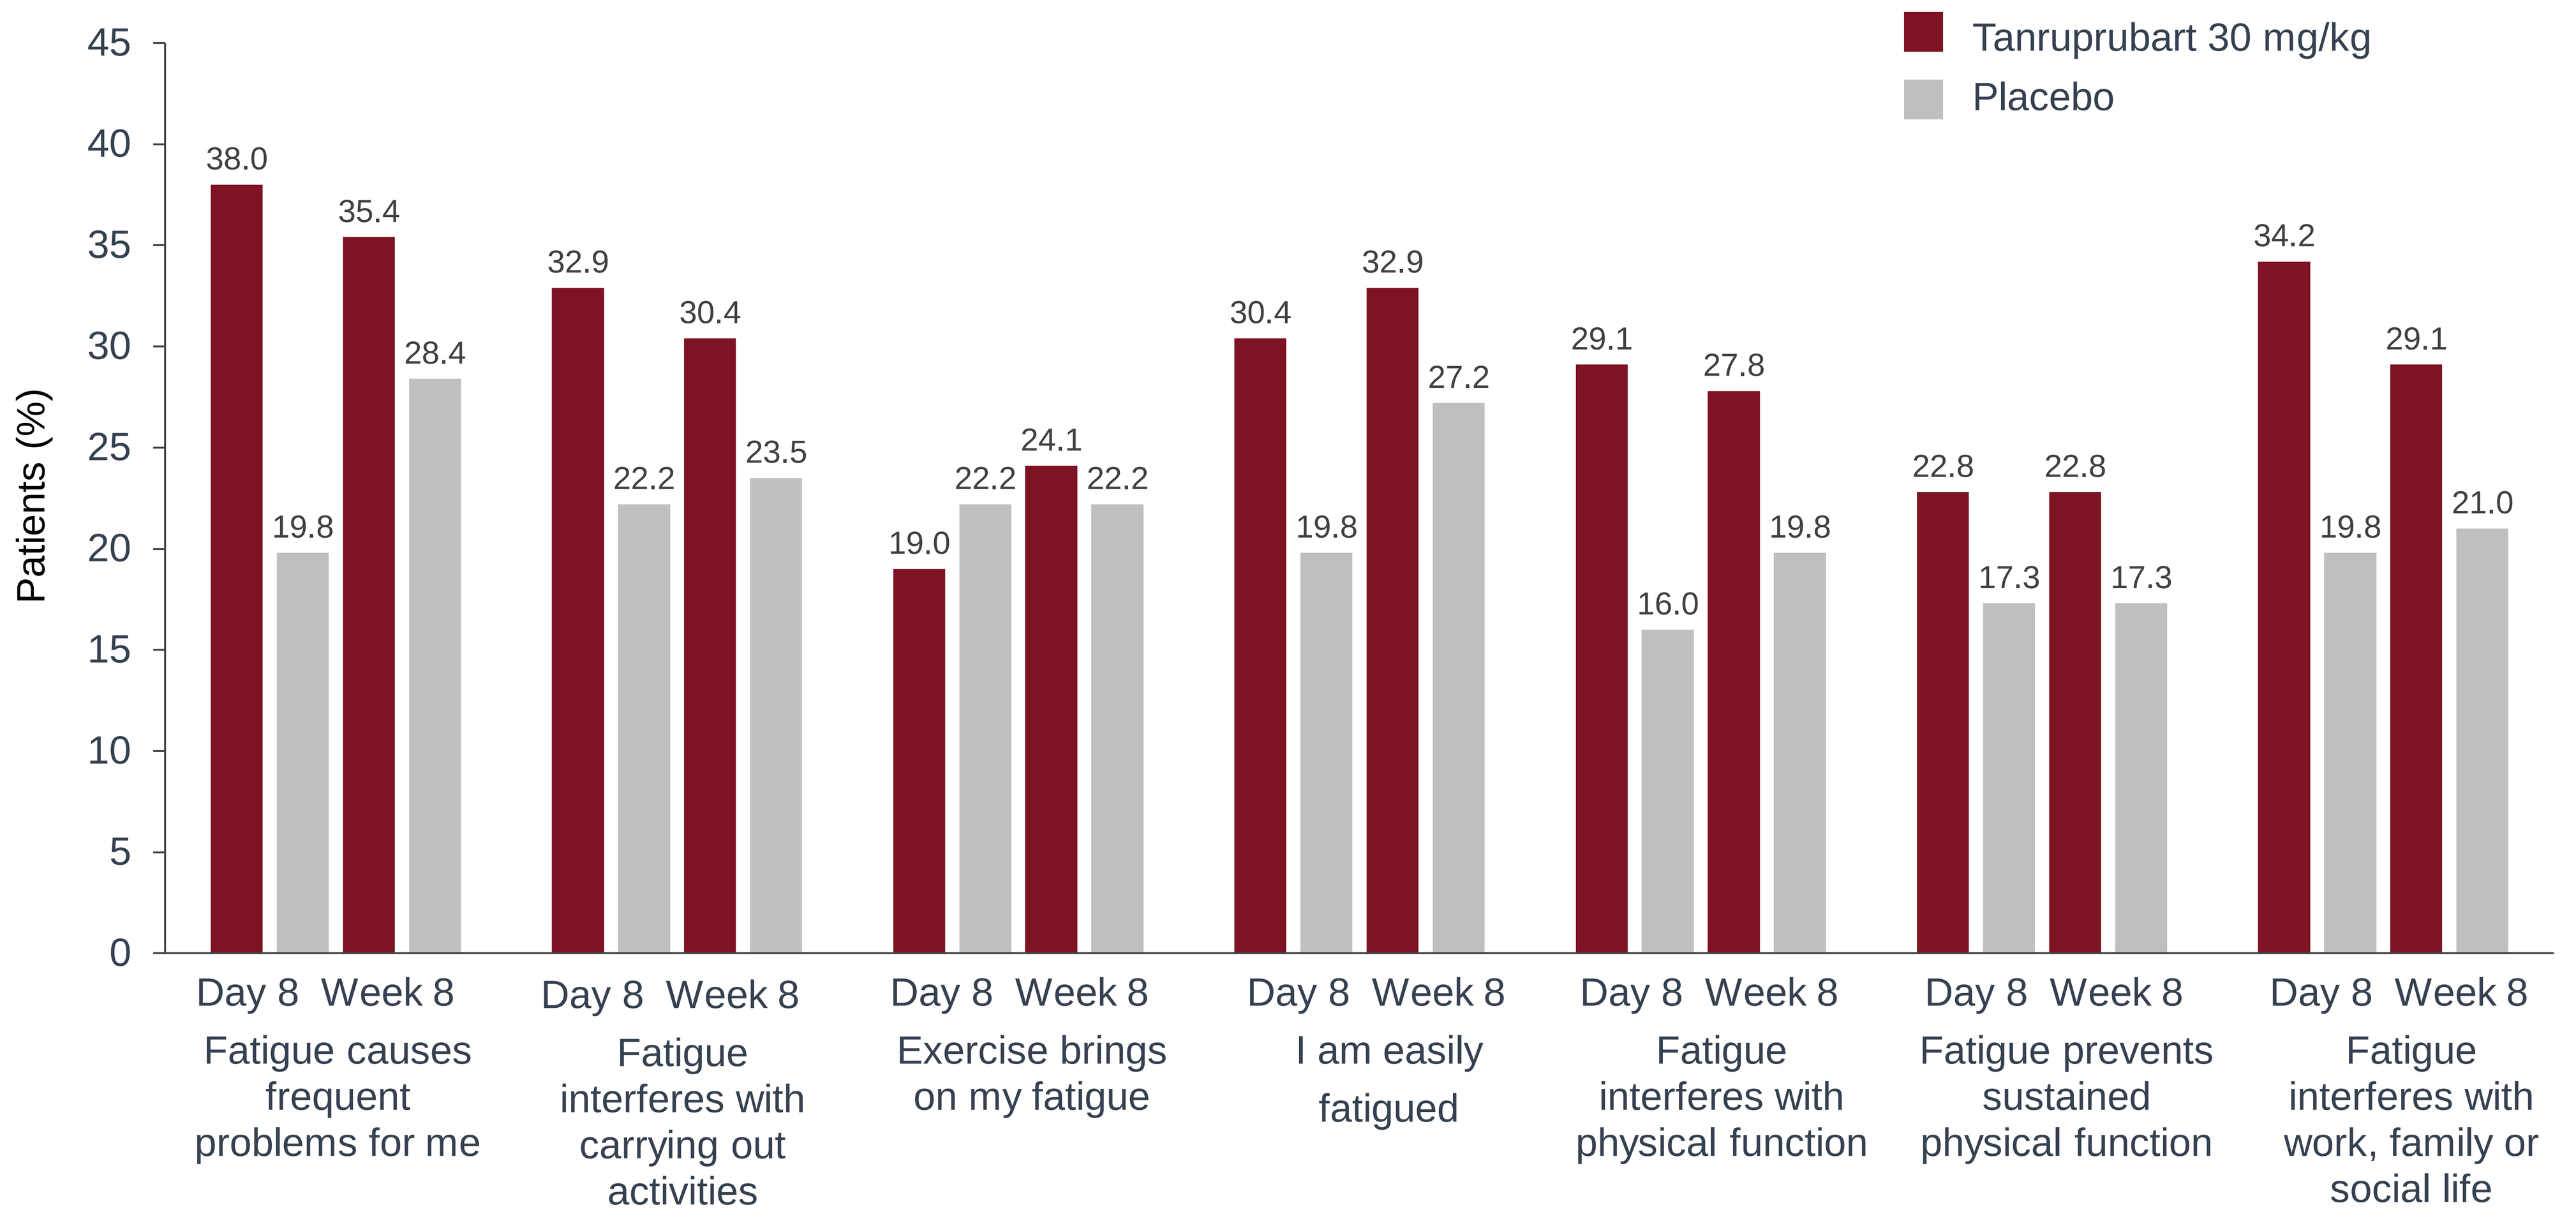

Tanruprubart 30 mg/kg
Placebo
Patients (%)
Fatigue causes frequent problems for me: 38.0, 19.8, 35.4, 28.4
Fatigue interferes with carrying out activities: 32.9, 22.2, 30.4, 23.5
Exercise brings on my fatigue: 19.0, 22.2, 24.1, 22.2
I am easily fatigued: 30.4, 19.8, 32.9, 27.2
Fatigue interferes with physical function: 29.1, 16.0, 27.8, 19.8
Fatigue prevents sustained physical function: 22.8, 17.3, 22.8, 17.3
Fatigue interferes with work, family or social life: 34.2, 19.8, 29.1, 21.0
Day 8
Week 8

Figure 3. PGIC improvement at Day 8 and Week 8

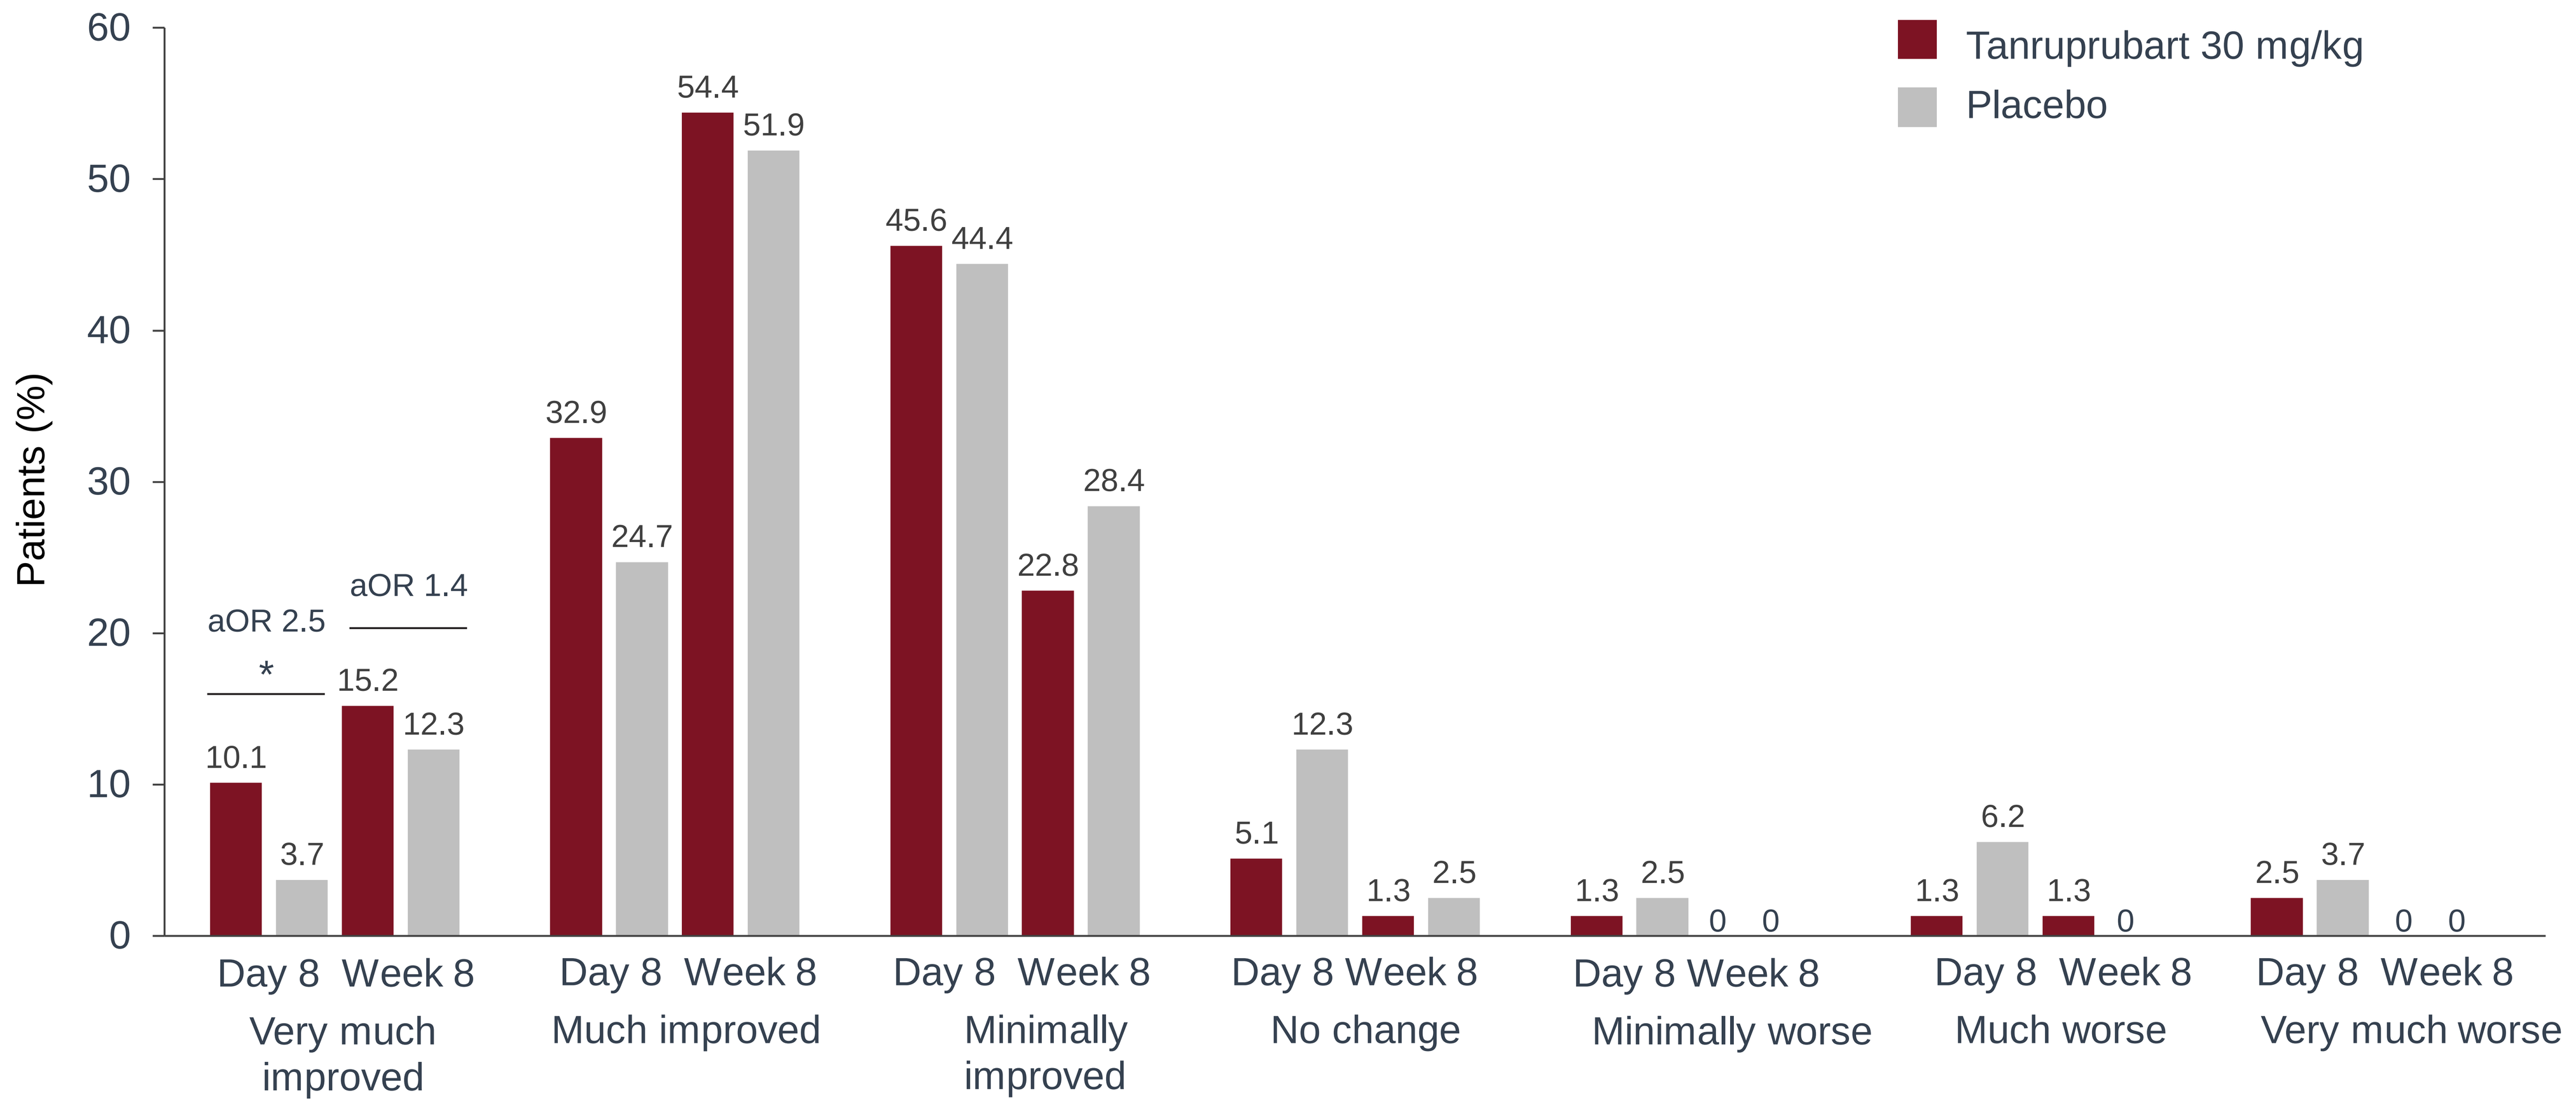

Tanruprubart 30 mg/kg
Placebo
Patients (%)
aOR 2.5 *
aOR 1.4
Very much improved: 10.1, 3.7, 15.2, 12.3
Much improved: 32.9, 24.7, 54.4, 51.9
Minimally improved: 45.6, 44.4, 22.8, 28.4
No change: 5.1, 12.3, 1.3, 2.5
Minimally worse: 1.3, 2.5, 0, 0
Much worse: 1.3, 6.2, 1.3, 0
Very much worse: 2.5, 3.7, 0, 0
Day 8
Week 8

*p<0.05.

CONCLUSIONS

- Tanruprubart demonstrated rapid and sustained improvements in health-related quality of life measures, consistent with observed objective clinical benefits
- These data further support the finding that classical complement inhibition with a single dose of tanruprubart rapidly reduces complement-mediated neuroinflammation and nerve damage leading to early stabilisation of patients during the acute progressive phase of GBS

References
1. Willison HJ, et al. *Lancet*. 2016;388:717–27. 2. van Doorn PA. *Presse Med*. 2013;42(6 Pt 2):e193–201. 3. van den Berg B, et al. *Nat Rev Neurol*. 2014;10:469–82. 4. Bernsen RA, et al. *Neurology*. 1999;53:409–10. 5. Lansita JA, et al. *Int J Toxicol*. 2017;36:449–62. 6. Suri P, et al. *Neurology*. 2022;98(18 Suppl):3867. 7. Kroon H-A, et al. Presented at the Neuromuscular Study Group Annual Scientific Meeting, September 20–22, 2022, Tarrytown, NY, USA.

Acknowledgments
The study was sponsored by Annexon Biosciences (Brisbane, CA, USA). Medical writing and editing assistance were provided by Envision Ignite, an Envision Medical Communications agency, a part of Envision Pharma Group and were funded by Annexon Biosciences.

Disclosures
MK, GM, JD, H-AK: Employment with and shareholder of Annexon Biosciences. QDM: Consultancy/advisory role with Annexon Biosciences. NP: No relevant disclosures. KCG: Consultancy/advisory role with Annexon Biosciences, argenx, Janssen, and Sanofi.
For additional information, please contact Myoung Kim: mkim@annexonbio.com.

Presented at the 12th Congress of the European Academy of Neurology (EAN); 27–30 June 2026; Geneva, Switzerland